13th World Congress of Cosmetic Dermatology, June 28 - July 1, 2018, Dubrovnik

Dr Irena Eris
COSMETIC LABORATORIES

Two novel vitamin D precursors in skin photoprotection

Bożena Tyszczuk[1], Monika Pasikowska-Piwko[1], Renata Dębowska[1], Katarzyna Równy[1], Carmen Vincent[2], Beata Ostrowska[1], Konrad Zieliński[3], Irena Eris[1]

[1]Dr Irena Eris Cosmetic Laboratories, R&D Department, Piaseczno, Poland, [2]Dermatology and Aesthetic Surgery, Private Practise, Piaseczno, Poland, [3]Pharmaceutical Research Institute, Warsaw, Poland

INTRODUCTION

Proper photoprotection is crucial in prevention of skin cancer, photoageing and hyperpigmentation. However, there is some controversy regarding sun induced vitamin D synthesis. It is claimed, that creams with high content of UV filters significantly inhibit vitamin D production in the epidermis.

AIM OF THE STUDY

The aim of the study was to evaluate and compare the efficacy of two cosmetic products for face photoprotection with SPF 50+. Each formulation contained a novel vitamin D precursor: CF-5 or C-9.

MATERIALS AND METHODS

In vitro test

In order to evaluate safety and efficacy of products (emulsion no. 14906 with two different vitamin D precursors: CF-5 and C-9, patent pending) we performed in vitro tests: cytotoxicity on L929 cells according to ISO 10993-5:2009; 3T3 NRU phototoxicity according OECD TG 432; and skin irritation on EpiDerm skin model according to OECD test guideline 439.

In vivo test

In vivo tests were performed in three groups of volunteers: 13 adults, 13 children (1-11 y. o.) and 20 adults after needle mesotherapy or anti-acne treatment with isotretinoin used orally. They were simultaneously testing both emulsions in a split-face test for 2 weeks. At the end of the test self-evaluation as well as a clinical assessment (according to analogous scale) were performed. Additionally 5 adult volunteers had instrumental measurements after 1, 2, 3 and 5 hours after creams application on the inner part of forearm: skin moisture (using Corneometer ® CM 825), skin anointment (using Sebumeter ® SM 815), transepidermal water loss (using Tewameter ® TM 300), skin pH (pH-Meter ® PH 905) and roughness of the skin surface (using Visioscan ®VC 98USB).

RESULTS

Results *IN VITRO*

Both creams were non-cytotoxic at the concentration of at least or equal to 0,001% (Fig. 1), but emulsion with CF-5 had less irritant potential.

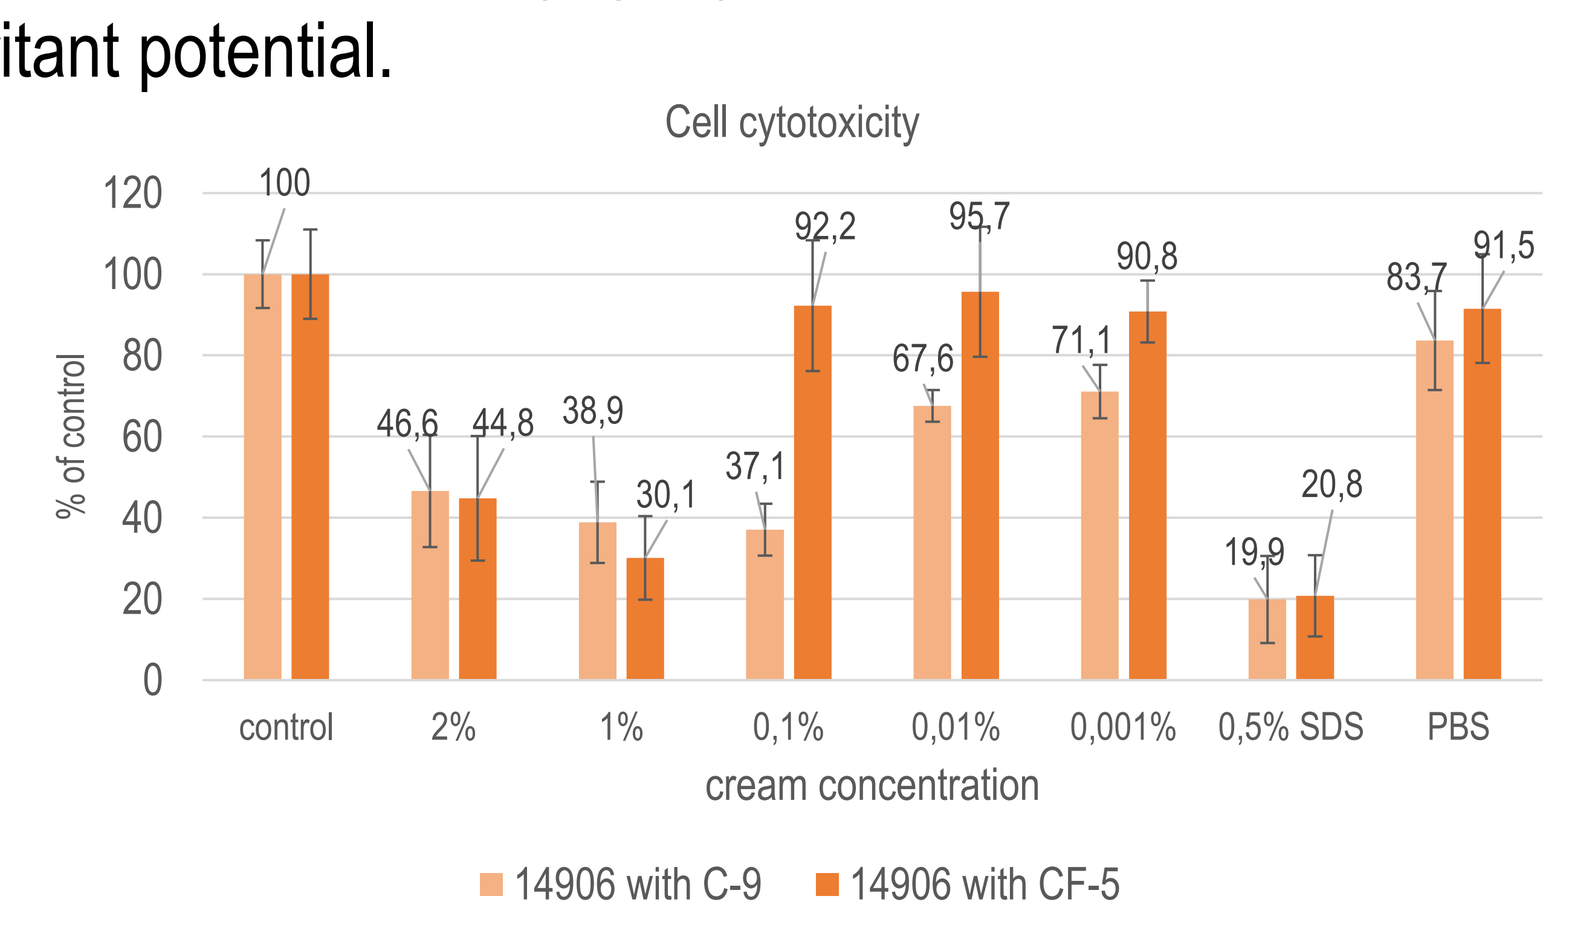
Cell cytotoxicity
% of control
control
2%
1%
0,1%
0,01%
0,001%
0,5% SDS
PBS
cream concentration
14906 with C-9
14906 with CF-5

Figure 1. MTT cytotoxicity test on L929 cells of emulsions no. 14906 with CF-5 or C-9 according to ISO 10993-5:2009. The product is non-cytotoxic when its viability is >70% of control at least in one concentration tested. Cream 14906 with CF-5 is non-cytotoxic at the concentration less than or equal to 0,1%, and 14906 with C-9 is non-cytotoxic at the concentration less than or equal to 0,001%.

The emulsion with CF-5 had no irritating potential (Fig. 2) after UV-B radiation.

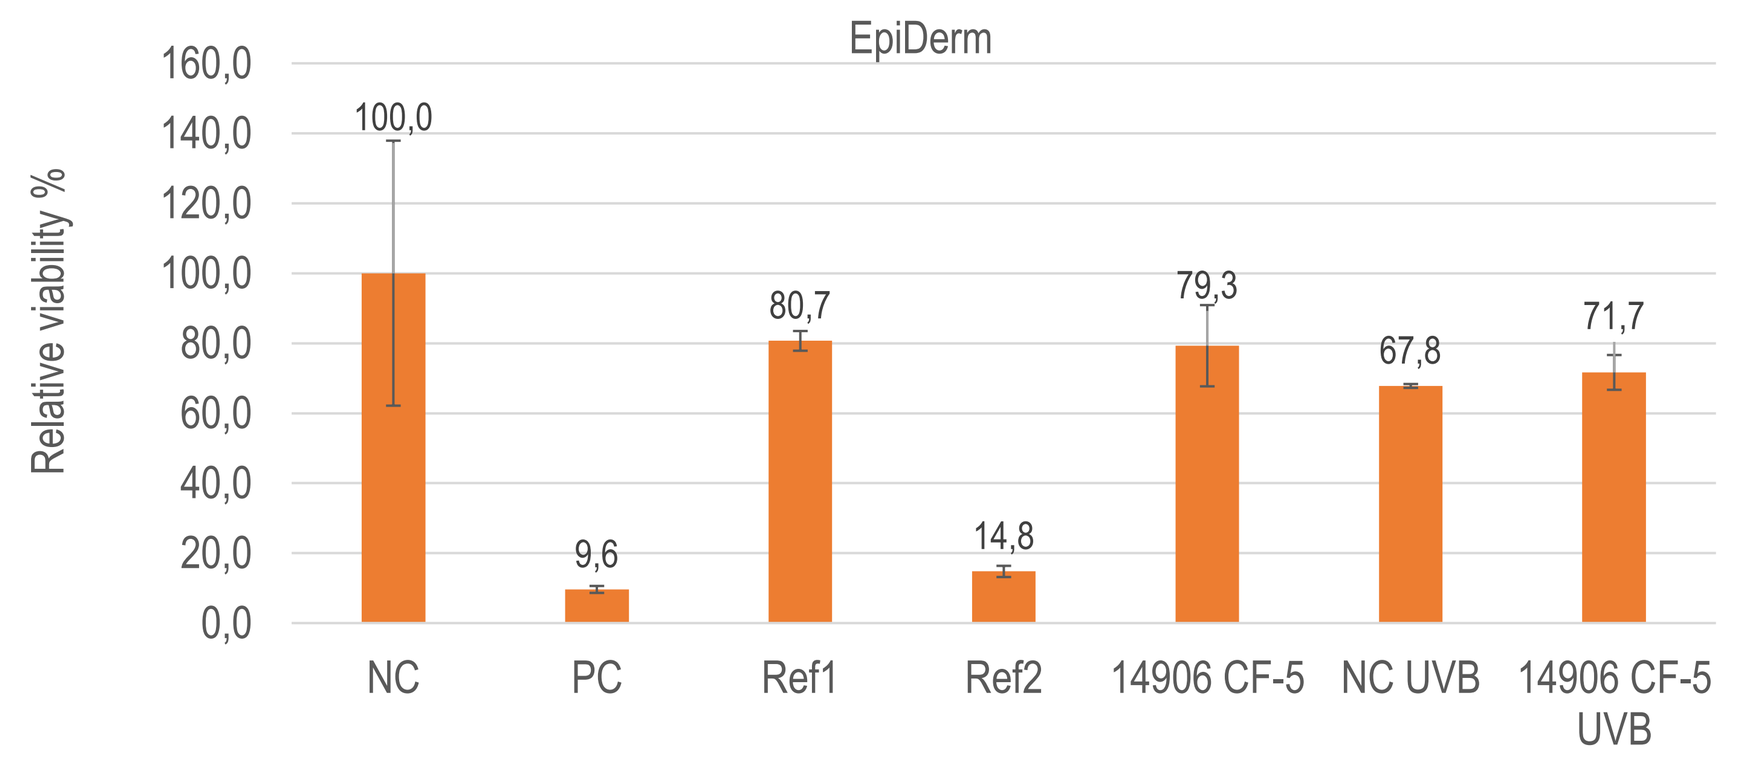
EpiDerm
Relative viability %
NC
PC
Ref1
Ref2
14906 CF-5
NC UVB
14906 CF-5 UVB

Figure 2. Skin irritation on EpiDerm skin model. An irritant is predicted if the mean relative tissue viability of three individual tissues exposed to the test substance is reduced below 50% of the mean viability of the negative controls. 14906 with CF-5 is non-irritant and non-phototoxic after UV-B radiation. PC – 5% SDS, Ref 1 - naphthalene acetic acid , Ref 2 - cyclamen aldehyde, NC – negative control.

The test results (MPE value in Run 1 = 0.102 and PIF values 2.924 and 2.698) suggested that the CF-5 emulsion had probable phototoxic potential (in tested range of concencentration, Table 1).

Table. 1. 3T3 phototoxicity test results for CF-5. Based on the validation study, a test substance with a PIF < 2 or an MPE < 0,1 predicts: 'no phototoxicity'. A PIF > 2 and < 5 or an MPE > 0,1 and < 0,15 predicts: 'probable phototoxicity'; and a PIF > 5 or an MPE > 0,15 predicts: 'phototoxicity'. Classification of phototoxic potential: probably photototoxic in concentrations: 4.64-1000 μg/ml.

	Run 1	Run 2
	IC50(+UV) = 56.5 μg/ml	IC_{50}(+UV) = 99.0 μg/ml
	IC50(-UV) = 163.8 μg/ml	IC_{50}(-UV) = 266.9 μg/ml
PIF	2.924	2.698
MPE	0.102	0.037

Results *IN VIVO*

The final concentration of CF-5 and C-9 precursors in the tested emulsions were 1μg/ml (lower than in the in vitro test - to avoid the probable UVA phototoxicity effect). Instrumental measurements (n=5) showed improvement of skin condition parameters. Moisturization increased by 26% for cream with C-9 precursor and by 22% for CF-5 precursor (4h after application, Fig. 3). We also observed improvement in skin ointment: measurement taken 4h after application of both products showed 12-times (C-9 precursor) and 4-times (CF-5 precursor) increase in skin anointment (Fig. 4). General skin condition (NRJ) measured 5h after products application showed improvement by 42% and 35% respectively for cream with C-9 and CF-5 precursors (data not shown). Moreover the number of wrinkles (Sew parameter) decreases 31% for product with C-9 precursor and 43% for cream with CF-5 precursor (measurements were taken 3h after application of products, Fig. 5). The test showed also TEWL reduction by 15% after use of cream with CF-5 precursor and by 6% after application of the cream with C-9 precursor (Fig. 6). Instrumental skin analysis evaluated by pHmeter reveal, that both tested emulsions had no negative impact on the physiological level of skin pH value (data not shown).

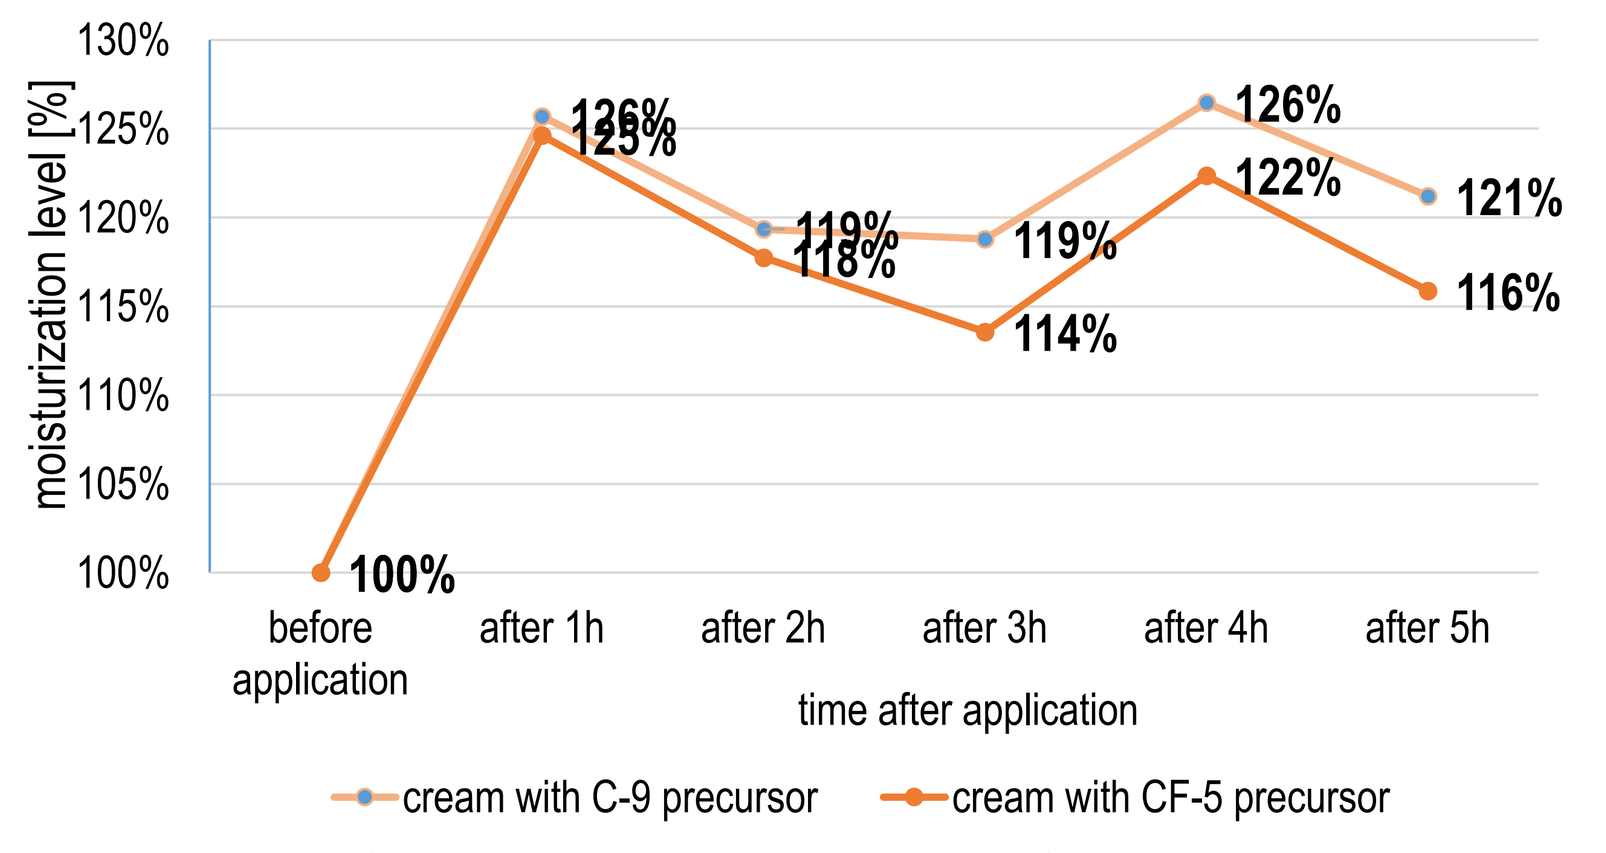
moisturization level [%]
before application
after 1h
after 2h
after 3h
after 4h
after 5h
time after application
cream with C-9 precursor
cream with CF-5 precursor

Fig.3. Changes in skin moisturization after single application of C-9 and CF-5 products.

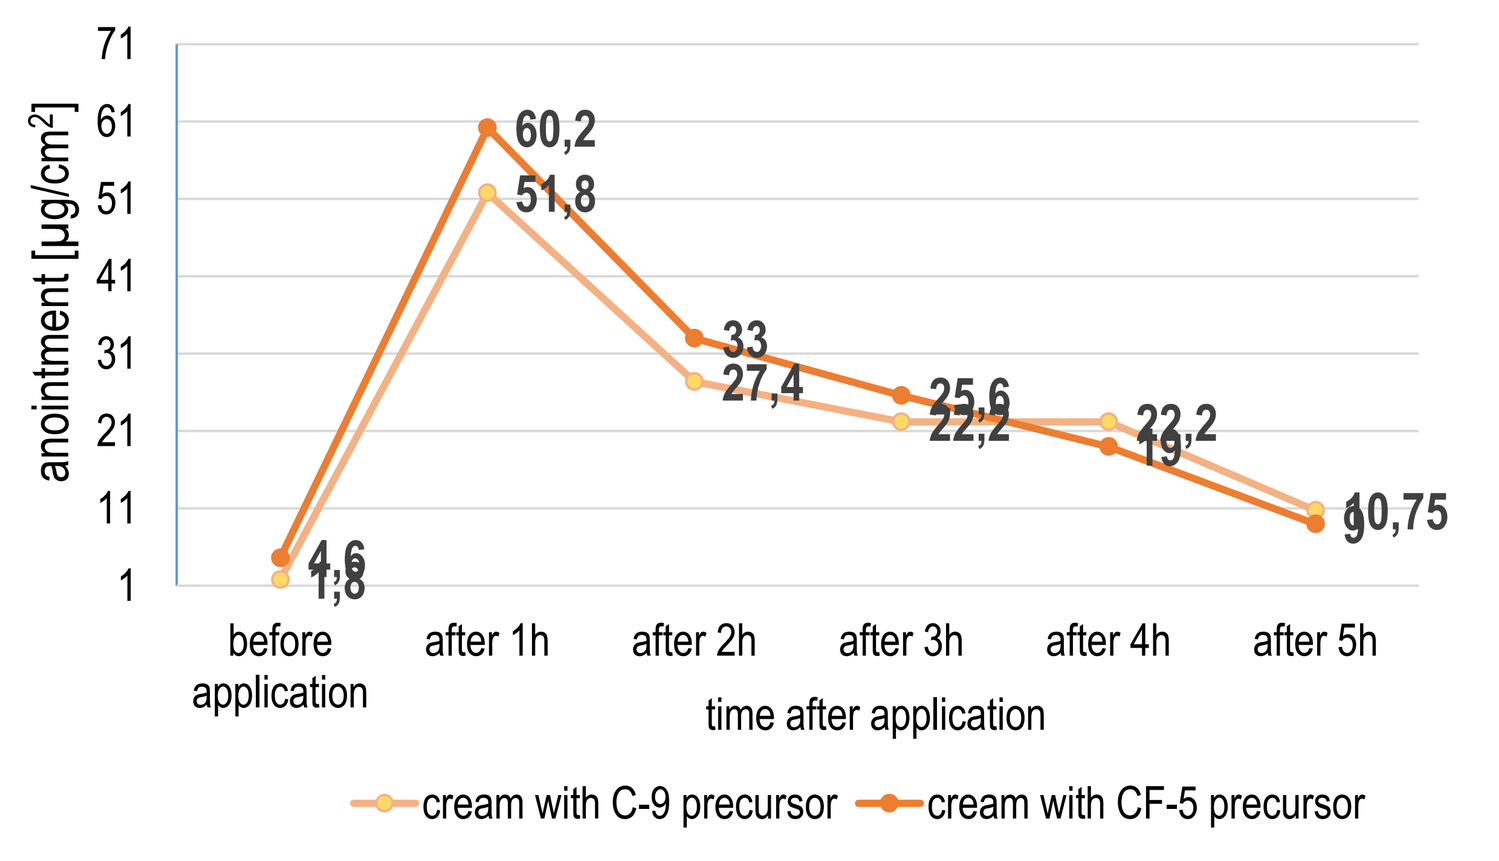
anointment [μg/cm2]
before application
after 1h
after 2h
after 3h
after 4h
after 5h
time after application
cream with C-9 precursor
cream with CF-5 precursor

Fig.4. Changes in anointment after single application of C-9 and CF-5 products.

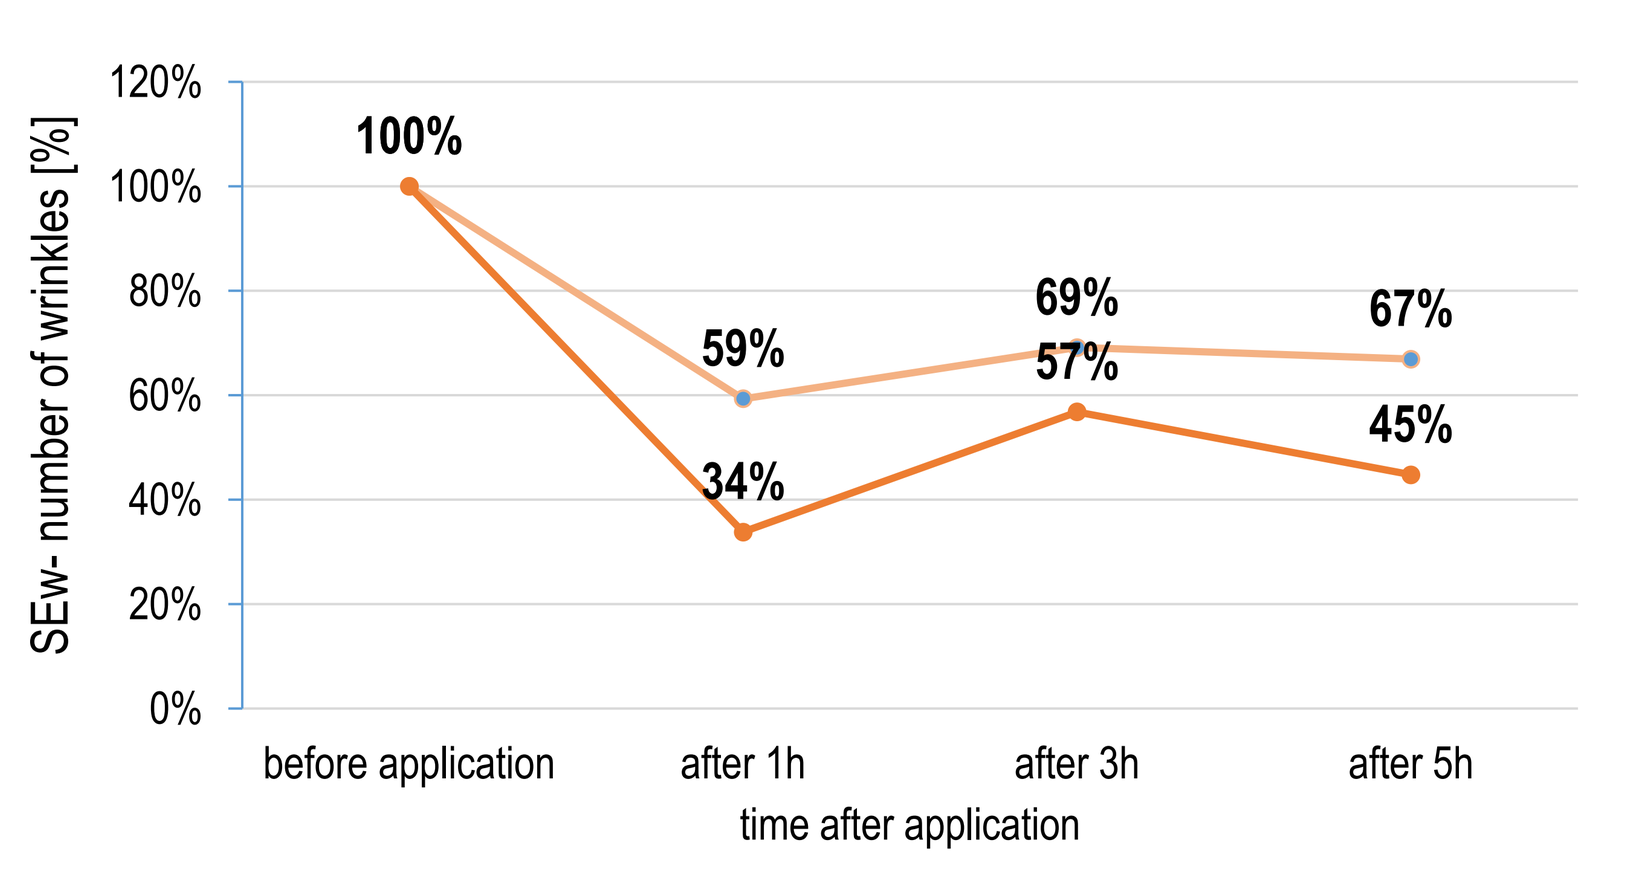
SEw- number of wrinkles [%]
before application
after 1h
after 3h
after 5h
time after application
cream with C-9 precursor
cream with CF-5 precursor

Fig. 5. Changes in number of wrinkles after application of C-9 and CF-5 products.

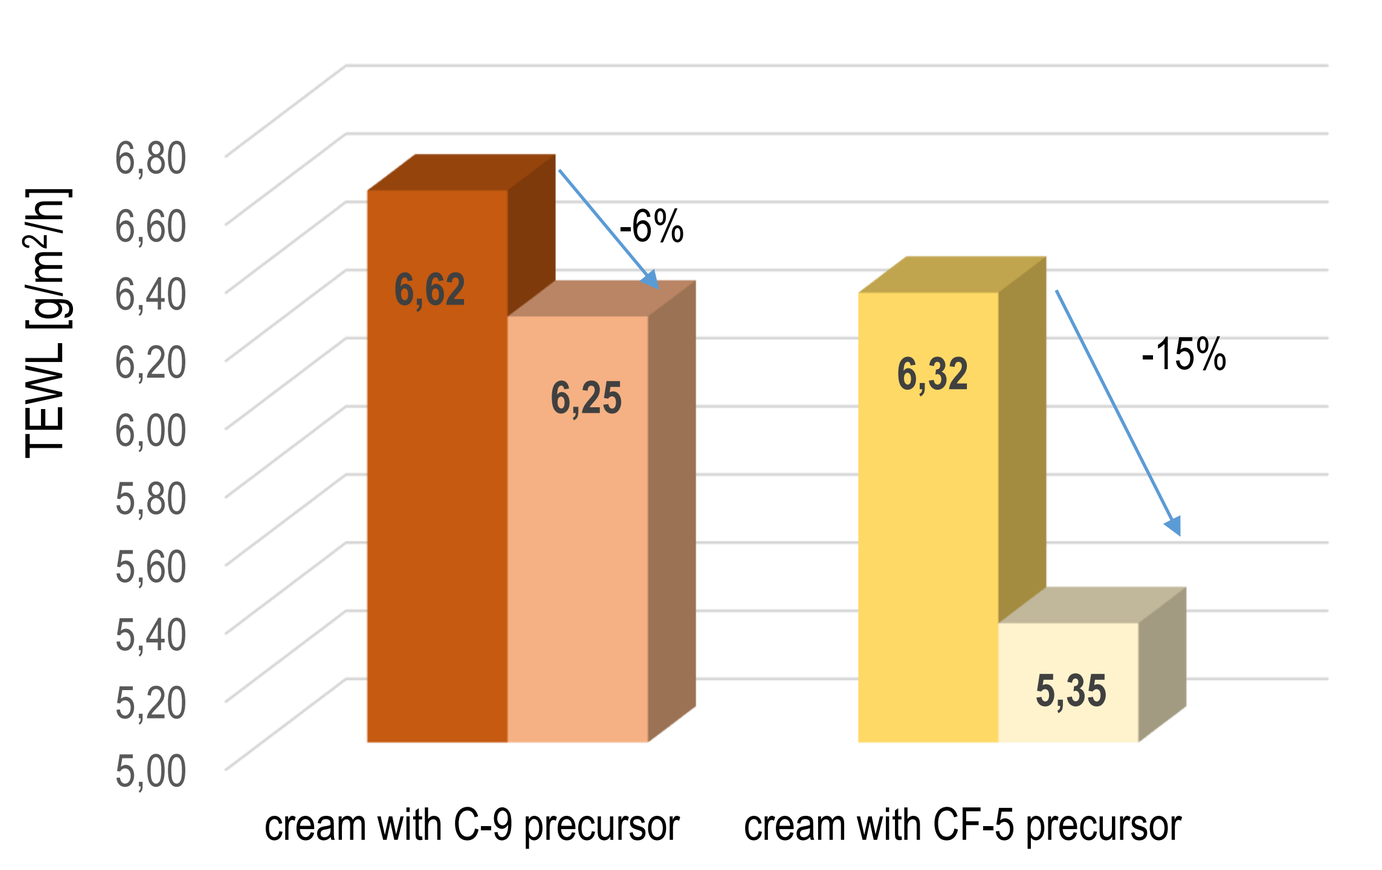
TEWL [g/m2/h]
6,62
6,25
-6%
6,32
5,35
-15%
cream with C-9 precursor
cream with CF-5 precursor

Fig. 6. Changes in TEWL after application of C-9 and CF-5 products.

Both creams were well tolerated when used after aesthetic medicine treatments (needle mesotherapy) as well as after dermatological treatment (oral isotretinoin). According to dermatological assessment in 10-point analogue scale, (where 10 means severe symptoms, 5 – average intensity and 1 – lack of symptoms) both tested products provide to 35% decrease in skin redness and 27% decrease in skin dryness (Tab. 2) for patients after retinoids treatment. In group of patients after mesotherapy dermatologists observed 57% decrease in skin redness and 35% decrease in skin dryness (Tab. 3). Self-evaluation provided by patients after 2 weeks of using both tested creams showed high positive impact of tested emulsions on skin condition (Tab. 4). Moreover, volunteers' evaluation (according to the same questionnaire survey as used in clinical trial) showed improvement in skin moisture, anointment and smoothness after both tested products usage (Tab. 5).

Table 2. Dermatological evaluation of patients skin condition before and after 7 days of product use according to 10-point analogue scale, where 10 means severe symptoms, 5 – average intensity and 1 – lack of symptoms.

Results for patients after retinoids treatment, n=15		
clinical evaluation of skin patient condition	D0	D7
skin dryness	8,9	6,5
skin redness	9,1	5,9

Table 3. Dermatological evaluation of patients skin condition before and after 7 days of product use according to 10-point analogue scale, where 10 means severe symptoms, 5 – average intensity and 1 – lack of symptoms.

Results for patients after mesotherapy, n= 5		
clinical evaluation of skin patient condition	D0	D7
skin dryness	6,8	4,4
skin redness	9,2	4,0

Table 4. Comparison of the efficacy of two tested products according to patients self-evaluation.

Effects seen after 2 weeks of treatment	**cream with C-9**	**cream with CF-5**	**Both creams (with C-9 and CF-5)**
patients after retinoid therapy, n= 15 [% of patients]			**patients after mesotherapy, n= 5 [% of patients]**
moisturizes the skin	80%	54%	60%
reduces skin dryness	66%	66%	80%
dries the skin	0%	7%	0%
nourishes the skin	27%	14%	40%
smoothes the skin	53%	53%	80%
the skin is regenerated	67%	47%	80%

Table 5. Results of self-evaluation of adults and children volunteers done after 2 weeks of products use.

	Effects seen after 2 weeks of usage	**cream with C-9 precursor [% of volunteers]**	**cream with CF-5 precursor [% of volunteers]**
adults, n=13	moisturizes the skin	77%	77%
	reduces skin dryness	69%	84%
	nourishes the skin	77%	69%
	smoothes the skin	77%	77%
children, n=13	reduces skin dryness	77%	77%
	smoothes the skin	77%	77%

CONCLUSION

It is known that vitamin D is important for the good skin condition. It takes part in healing processes and proper cell proliferation. Here it is presented for the first time the use of novel vitamin D precursors in photoprotective creams which are beneficial for skin condition also after different aesthetic medicine treatments.

This work was supported by Mazowiecka Jednostka Wdrażania Programów Unijnych grant no. RPMA.01.02.00-14-5671/16-00