29-th EADV Congress, 28.10 – 01.11.2020, Vienna

The use of combination of potassium azeloyl diglycinate, shikimic acid and niacinamide in topical application on skin with comedonal acne.

Tyszczuk B.[1], Kuranc A.[1], Koziej P. [2] , Koziej J.[2], Dębowska R.[1], Rogiewicz K.[1], Eris I.[1]
[1]Dr Irena Eris Cosmetic Laboratories, R&D Department, Piaseczno, Poland [2]Dr Koziej Instytut Badań Kosmetyków, Warsaw, Poland

Introduction

Acne vulgaris is a common chronic skin condition. It is characterized by inflammation of sebaceous glands and hair follicles. Epidermal keratinization and seborrhea disorders are mainly caused by hormonal imbalance. Excessive sebum and the residue of dead skin cells accumulate in pilosebaceous units, which results in formation of whiteheads and blackheads. This creates a good environment to grow *Cutibacetirum acnes*, which may lead to inflammatory changes. Lesions frequently result in acne scars or post-inflammatory discolorations.

The aim of the study was to determine the effectiveness of cosmetic formulation (emulsion no. 14010) containing potassium azeloyl diglycinate (1.5%), shikimic acid (1.5%) and niacyniamide (1%) in patients with combination skin and with comedonal acne.

Methods

In order to evaluate safety and efficacy of the tested product, we performed *in vitro* skin irritation tests on EpiDerm skin model according to OECD test guideline 439 and ISO 10993-10:2013.
Skin patch test on 30 subjects with allergy-prone skin was performed to confirm dermatological safety of the emulsion.
The antioxidant activity (Radical Protection Factor - RPF) of the tested emulsion was determined using the 2.2-diphenyl-1-picrylhydrazyl (DPPH) radical scavenging method.
Determination of antibacterial properties of the tested product on *Cutibacterium acne* (ATCC 11827) and on total number of aerobic microorganisms isolated from the surface of acne lesions were done using the suspension method *in vitro*. Measurements were carried out at the baseline, and then after 30 minutes, 1, 2 and 3 hours.
The *in vivo* study was performed in group of 30 participants with oily, mixed and comedonal acne skin. The participants were asked to use the product two times a day for 3 weeks. Skin evaluation was done at the baseline and after 3 weeks of product application. Changes in sebum secretion (CK Multiskincenter 750), pores (VisioFaceQuick) and skin texture (MiravexAntera) were evaluated. All the participants completed a satisfaction questionnaire after 3 weeks of product usage.

Conclusion

In vitro experiments confirmed that the tested emulsion has no irritant potential and contains anti-ROS activity compounds. We confirmed its antibacterial activity. Dermatological patch test study indicated that the tested emulsion did not show any irritating or sensitizing properties. It is also highly effective in short time of application. The product was proved to have anti-acne efficacy and thus can be recommended for daily use on acne-prone skin, especially for comedonal acne.

Results

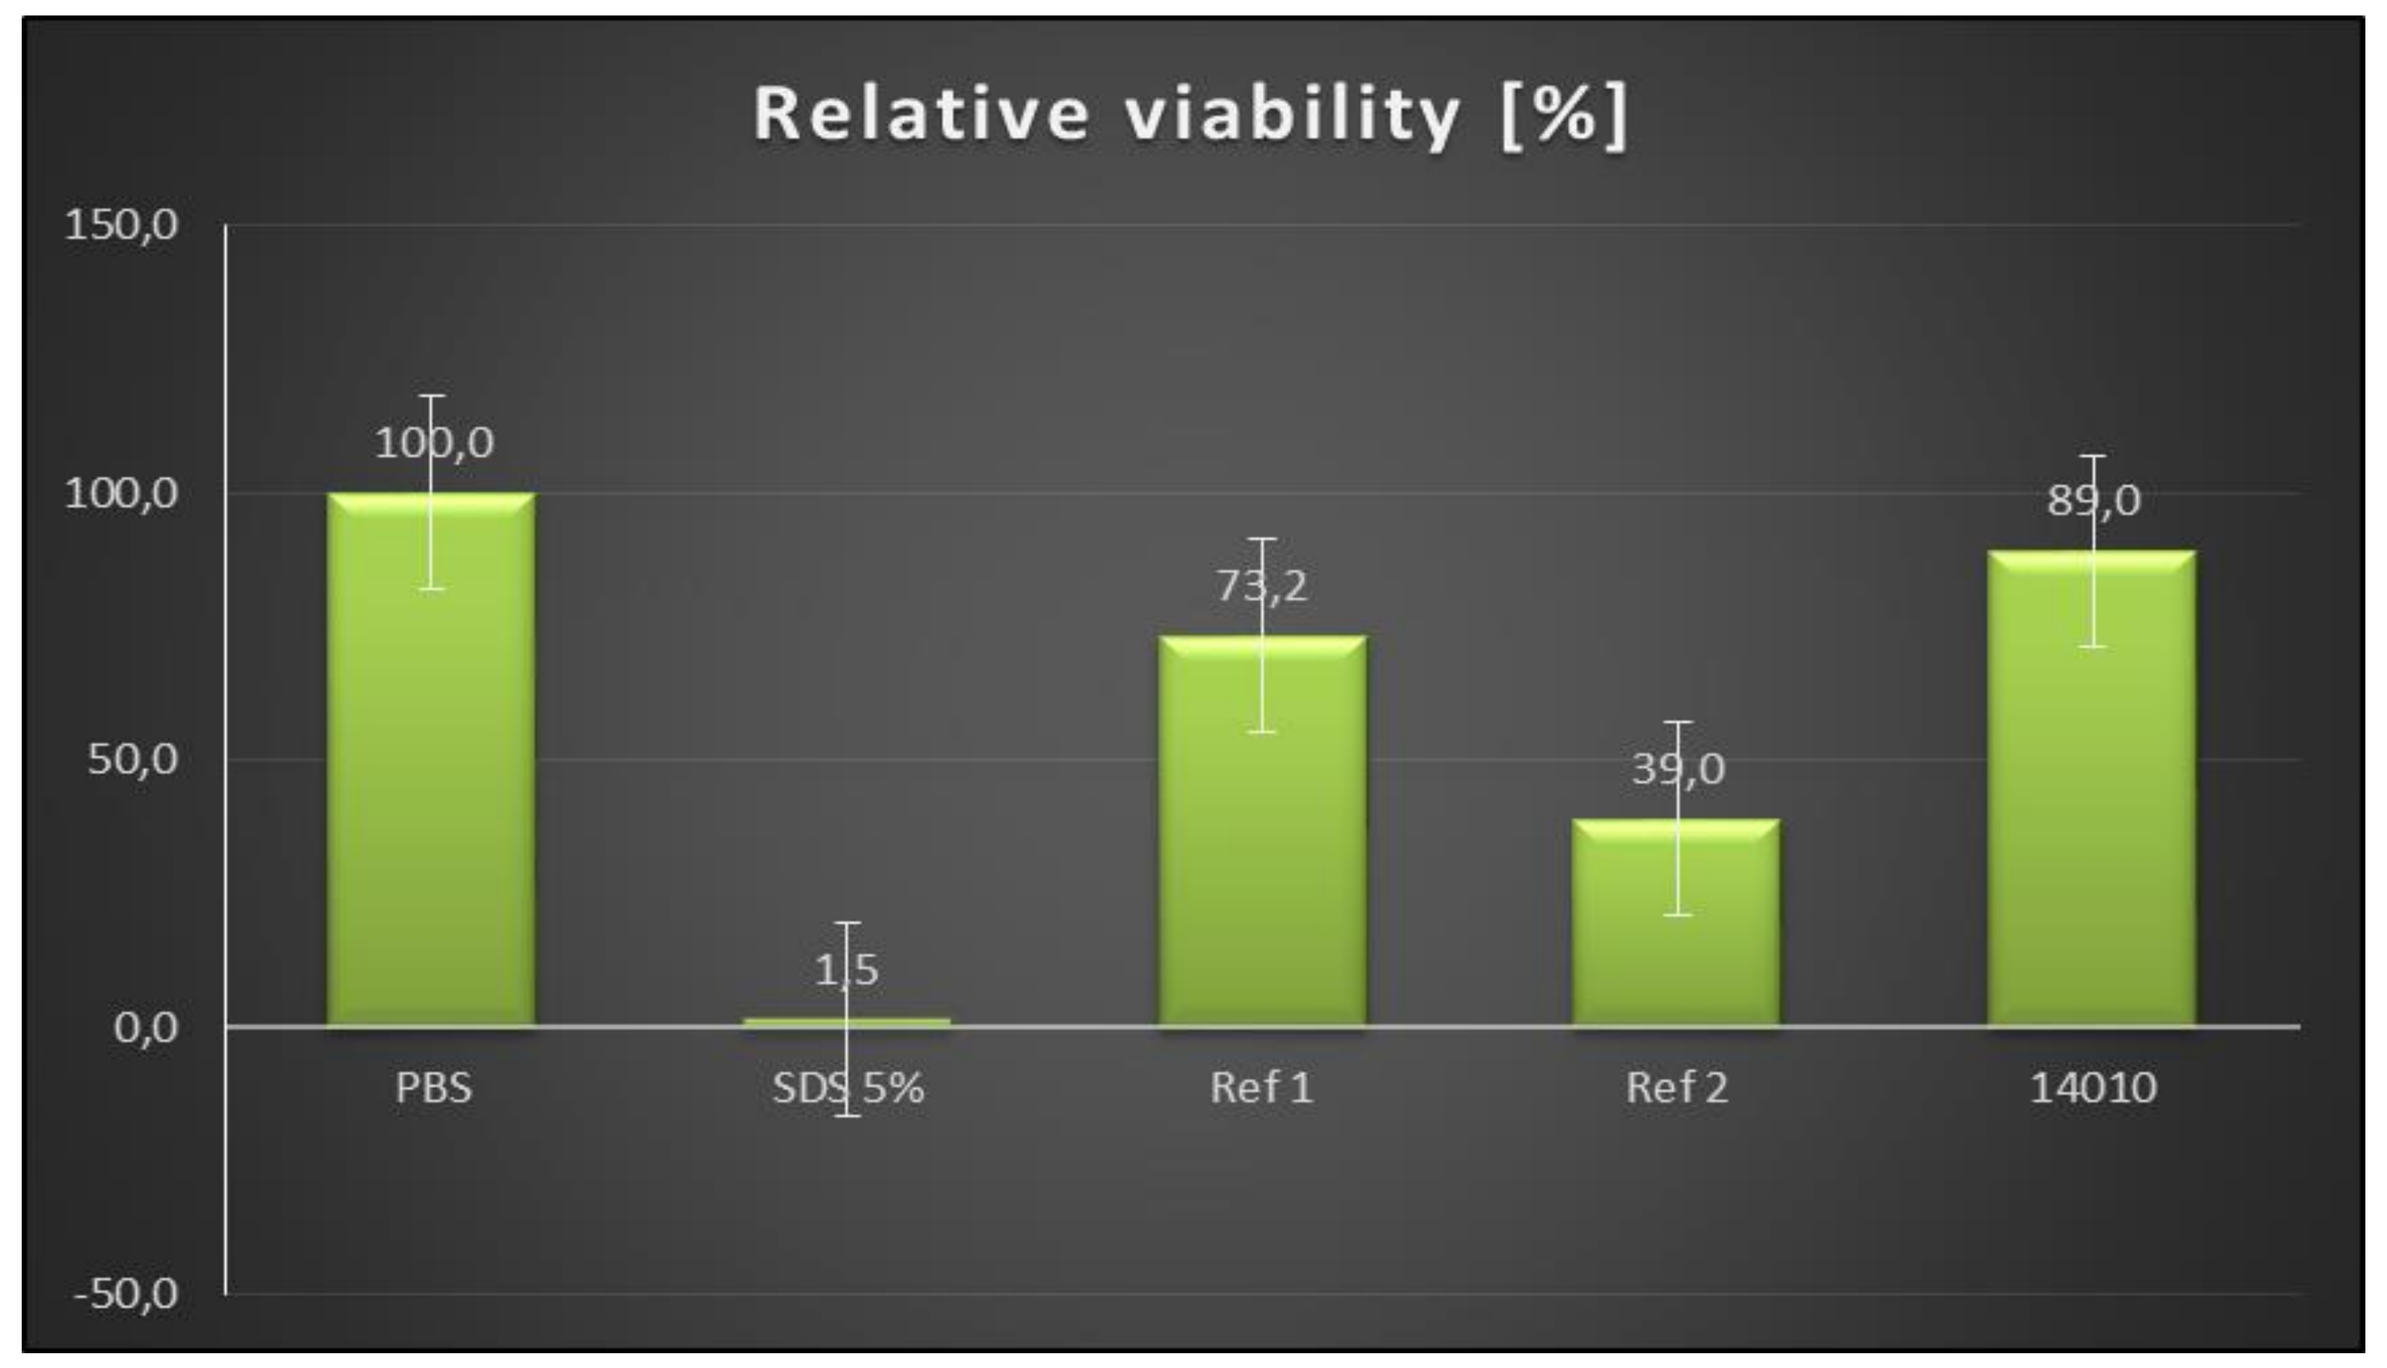
Relative viability [%]
150,0
100,0
50,0
0,0
-50,0
100,0
1,5
73,2
39,0
89,0
PBS
SDS 5%
Ref 1
Ref 2
14010

Figure 1. Skin irritation potential of the emulsion 14010 tested on EpiDerm model. Ref 1- naphthalene acetic acid (CAS 86-87-3) – non classified (non irritant). Ref 2 - cyclamen aldehyde (CAS 103-95-7) – classified (irritant, Cat. 2). Correlation of *in vitro* and *in vivo* results: Tissue viability ≤ 50% of the control (PBS) — irritant. Tissue viability ≥ 50% of the control — non-irritant.
Emulsion 14010 was confirmed as non-irritant on EpiDerm skin model, resulting in the mean tissue viability of 89%.

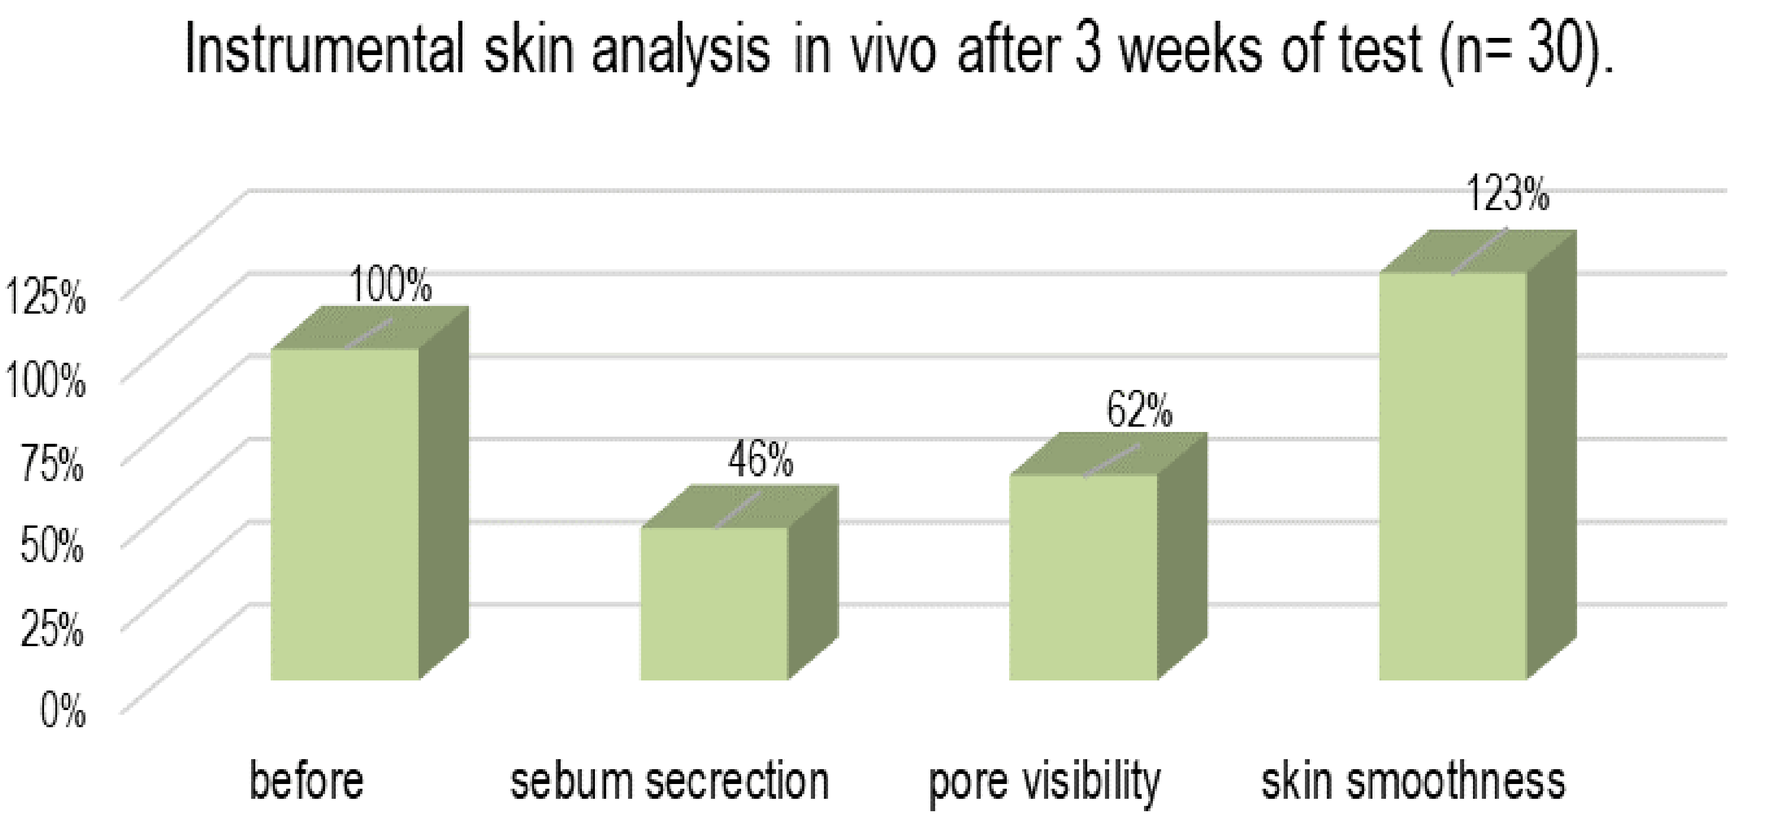
Instrumental skin analysis in vivo after 3 weeks of test (n= 30).
125%
100%
75%
50%
25%
0%
100%
46%
62%
123%
before
sebum secrection
pore visibility
skin smoothness

Figure 4. Instrumental skin analysis showed a decrease of sebum secretion by 54%, a reduction in pores visibility by 38% and an improvement of the skin smoothness by 23%.

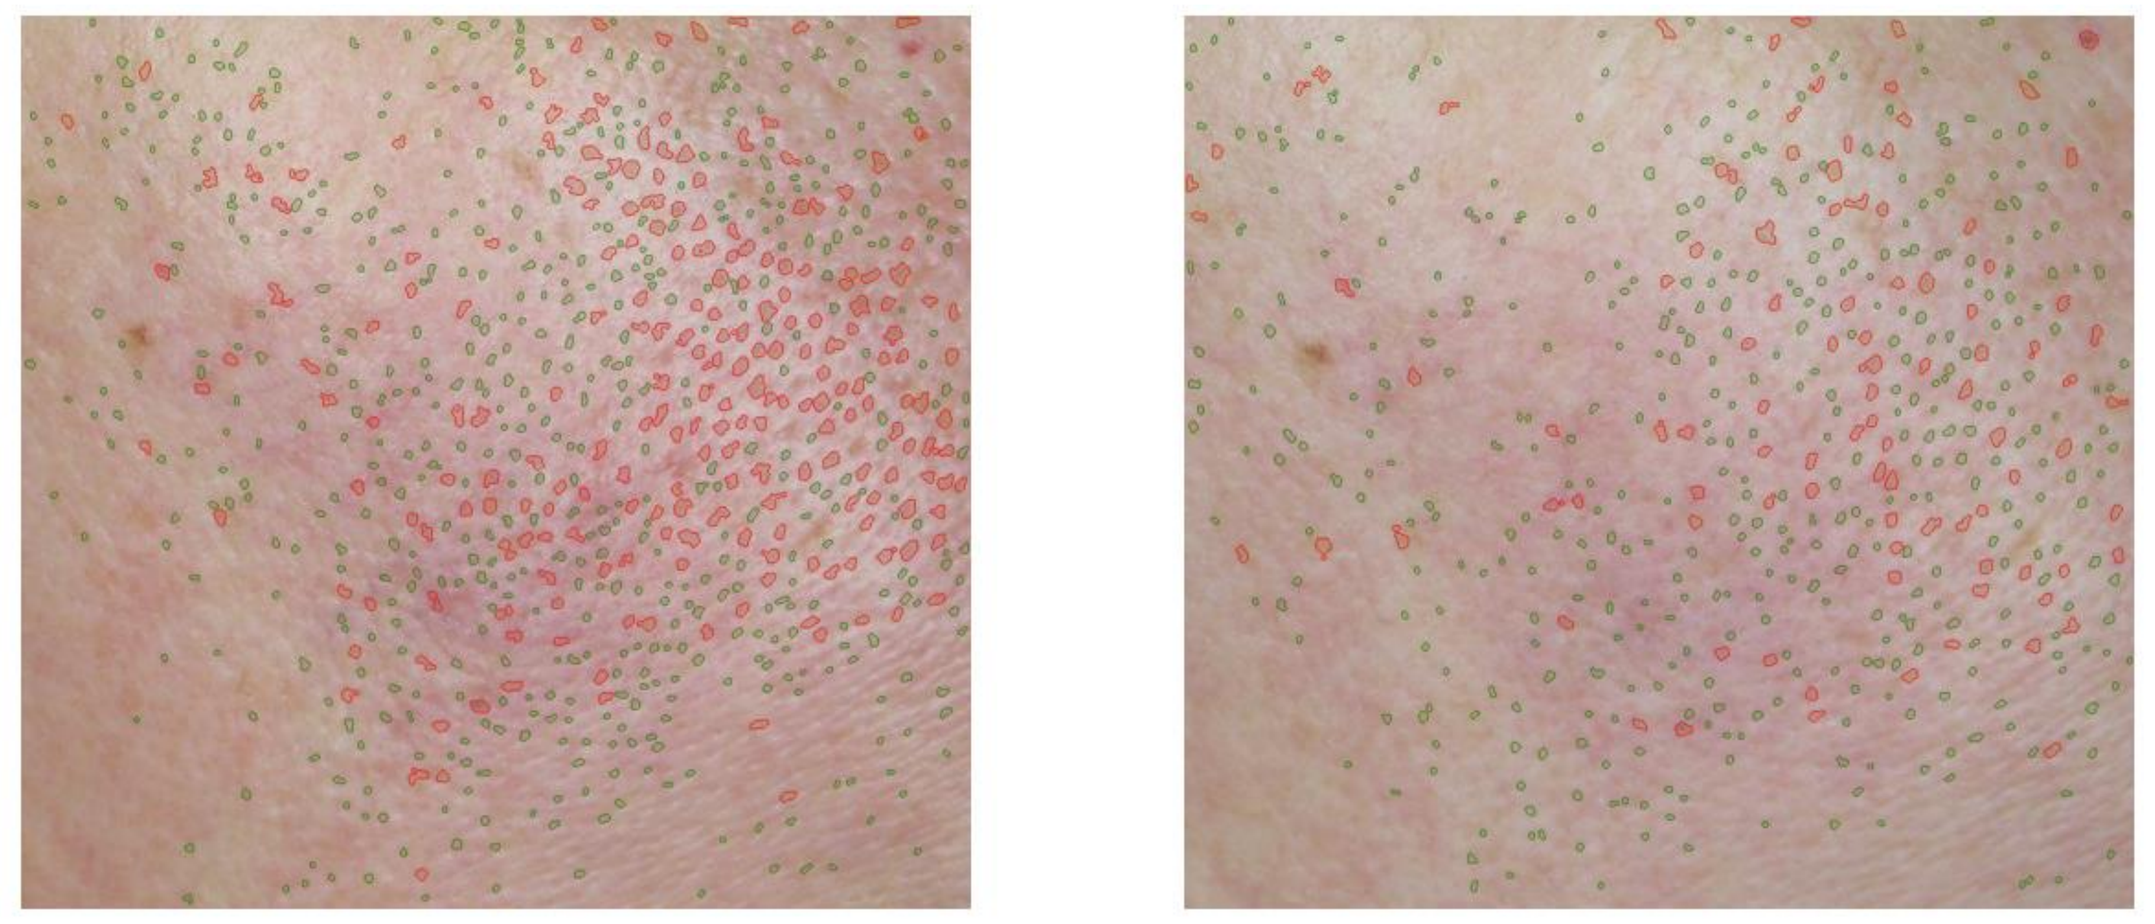

Figure 5. Top panel shows reduction of pores visibility by 32% after 3 weeks of tested product usage (VisioFaceQuick).

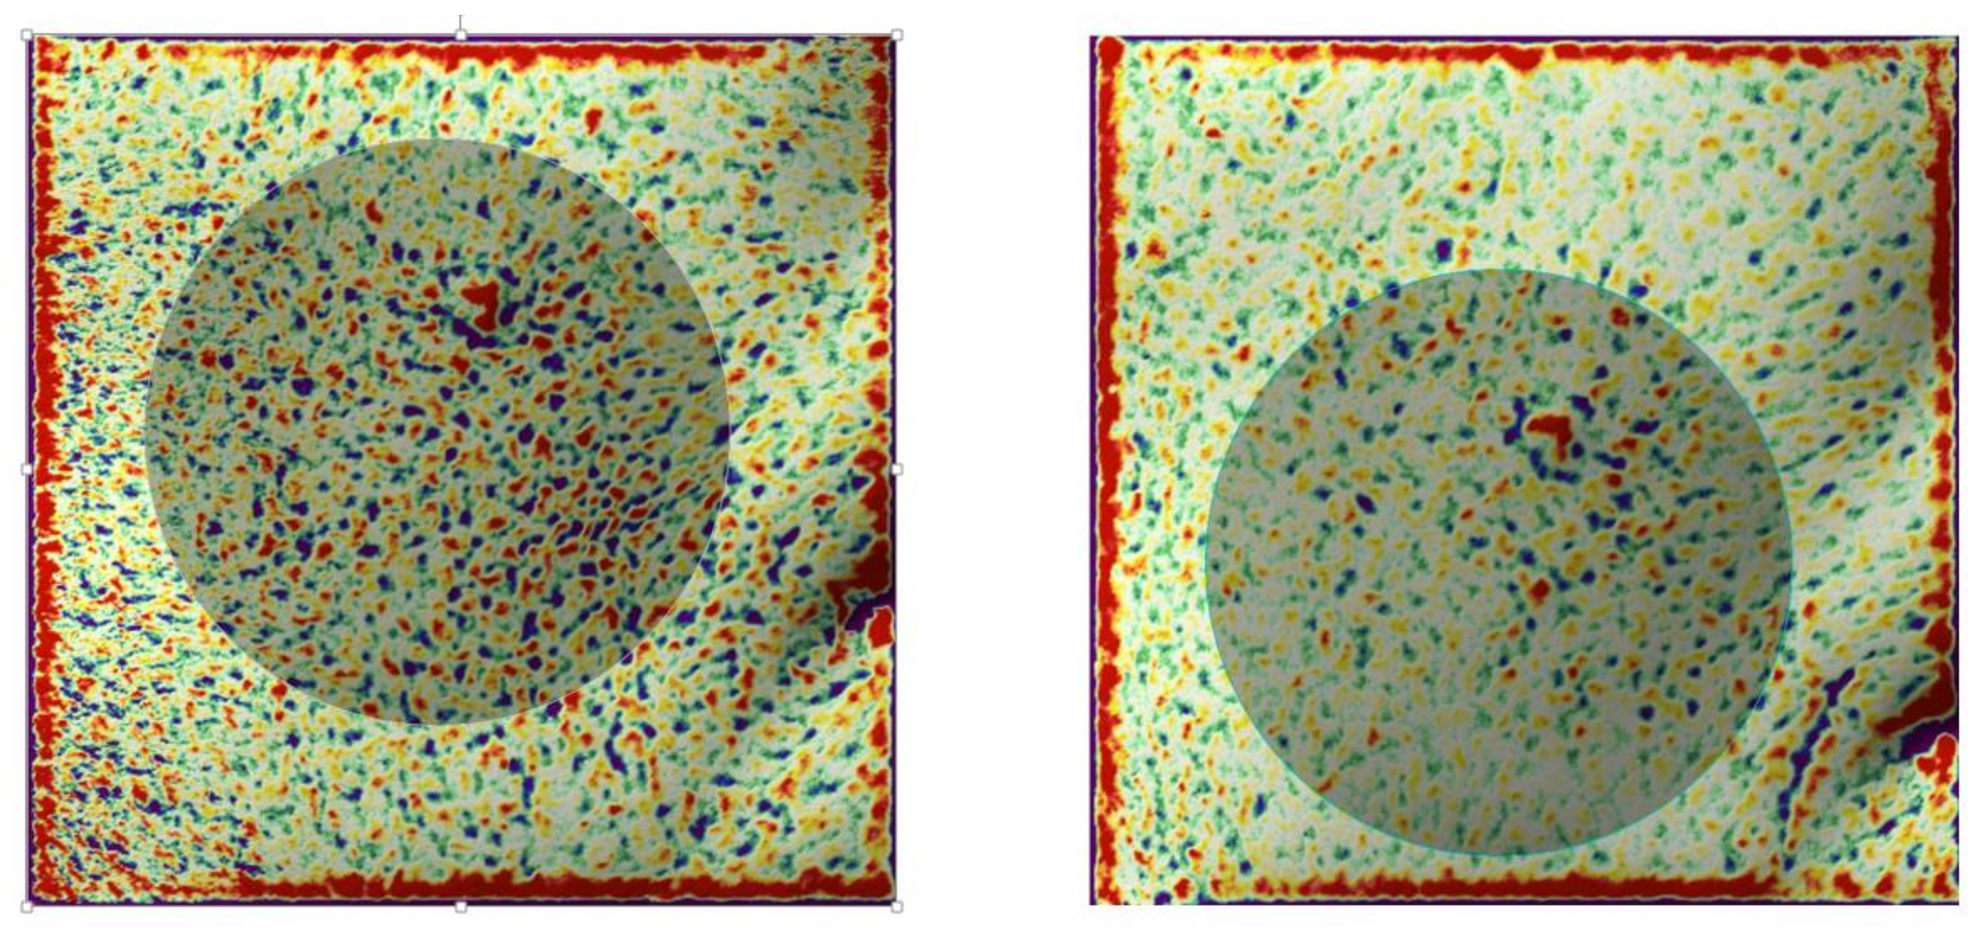

Figure 6. Best case report of skin texture smoothness improvement (+ 31%) after 3 weeks of emulsion 14010 usage (MiravexAntera).

	Antioxidant capacity (mg DPPH/ 1g sample)	RPF (10^{18} DPPH/ 1g sample)
14010	**1,06**	**1,72**

Figure 2. RPF value (Radical Protection Factor) was 1,72 x 10^{18} DPPH/1g of emulsion and antioxidant capacity was equal to 1,06 mg DPPH/1g of emulsion.

pathogens isolated from the acne lessions	after 30 min	after 1h	after 2h	after 3h
tested emulsion (14010) in concentration 100%	**94,80%**	>98,3%	>99,6%	>99,9
Cutibacterium acnes (ATCC 11827)	after 30 min	after 1h	after 2h	after 3h
tested emulsion (14010) in concentration 100%	**99,8%**	99,9%	>99,9%	>99,9%

Figure 3. After 30 minutes of contact between the tested product and the pathogens isolated from the acne lesions, and with commercial *Cutibacterium acne* (ATCC 11827), we observed reduction in the total number of aerobic microorganisms by 94.8% and 99.8%, respectively. After 3 hours, the reduction increased to over 99.9%.

The patch test did not show any irritating or sensitizing properties of the tested cosmetic formulation (data not shown).

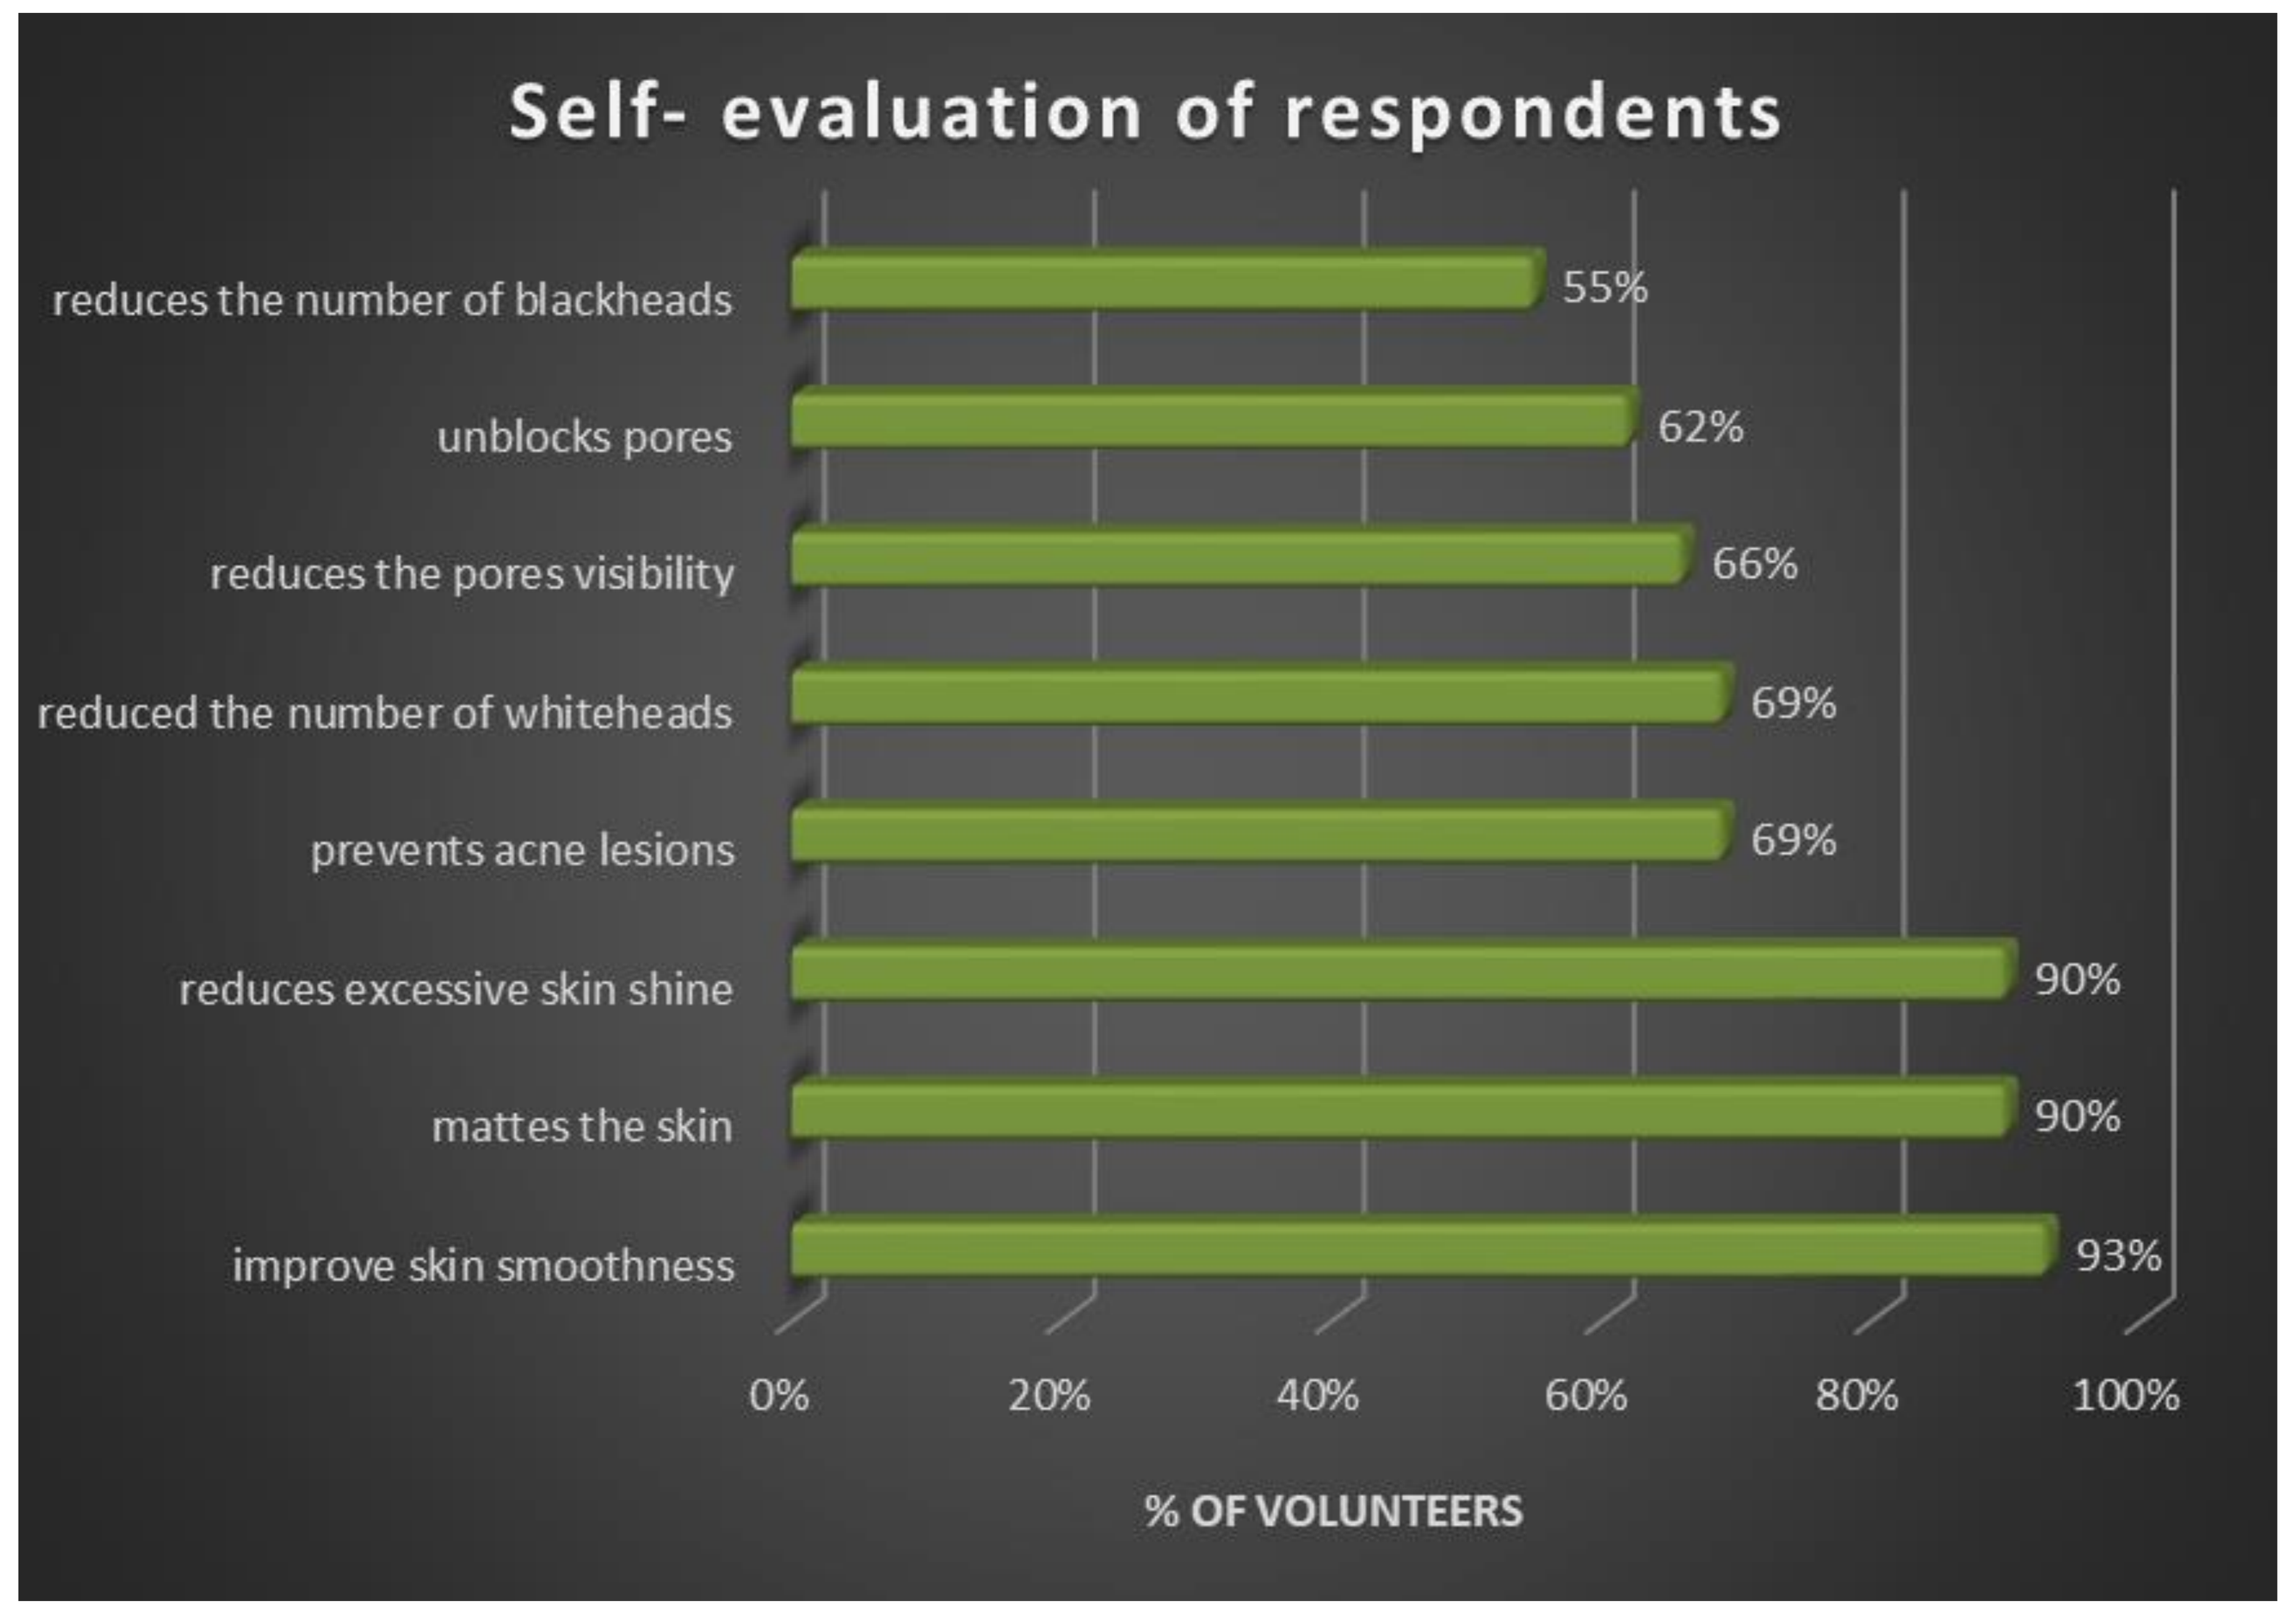
Self- evaluation of respondents
reduces the number of blackheads
55%
unblocks pores
62%
reduces the pores visibility
66%
reduced the number of whiteheads
69%
prevents acne lesions
69%
reduces excessive skin shine
90%
mattes the skin
90%
improve skin smoothness
93%
0%
20%
40%
60%
80%
100%
% OF VOLUNTEERS

Figure 7. Results of selected parameters after 3 weeks of the emulsion 14010 usage presented as a percentage (%) of the survey respondents who confirmed the benefits of the product.

In the subjective assessment 90% of volunteers reported a decrease in excessive skin shine and 93% patients observed improvement in skin smoothness. Moreover, volunteers confirmed that the tested product unblocked and reduced skin pores visibility. Furthermore, the participants reported, that blackheads appeared less visible and skin was matted.

This work was supported by Mazowiecka Jednostka Wdrażania Programów Unijnych grant no. RPMA.01.02.00-14-5671/16-00