49th Annual ESDR Meeting 2019, Bordeaux

Dr Irena Eris
COSMETIC LABORATORIES

Skin anti-ageing effects of mitochondrial potassium channels regulation by naringenin.

K. Śmigielska[1], K. Zdroik[1], A. Kołaczek[1], R. Dębowska[1], A. Szewczyk[2], P. Bednarczyk[3], K. Rogiewicz[1], I. Eris[1]

[1]Dr Irena Eris Cosmetic Laboratories, Piaseczno, Poland, [2]Nencki Institute of Experimental Biology, Polish Academy of Sciences, Warsaw, Poland [3]Warsaw University of Life Sciences - SGGW, Warsaw, Poland

INTRODUCTION

Skin ageing has been associated with a decline in mitochondrial function, increase in inflammatory biomarkers such as IL-6 and accumulation of damage caused by excessive reactive oxygen species. Activation of the mitochondrial potassium channels has been found to induce cytoprotective effects. The aim of the study was to assess a flavonoid, naringenin, as a topical ingredient targeting mitochondrial skin ageing *in vitro* and *in vivo*

MATERIALS AND METHODS

In vitro tests

Interaction of naringenin with ATP-regulated potassium channel (mitoK_{ATP}) and the mitochondrial large-conductance Ca^{2+}-regulated potassium channel (mitoBK_{Ca}) was tested in mitoplasts isolated from primary human dermal fibroblasts using patch-clamp technique (*Kampa et al., Exp Dermatol. 2019;00:1–9*). The effect on IL-6 level was assessed by ELISA method in human keratinocytes under UVB irradiation. Additionally SOD2 levels, activity of respiratory chain (Western Blot assay) and mitochondria morphology (confocal microscopy) following incubation with naringenin were evaluated in untreated and UVA-irradiated cells.

In vivo test

Emulsion no. 5552 containing naringenin was tested in vivo on 30 volunteers aged 39-69. Skin parameters were measured at baseline and after 4 weeks of use. Changes in hydration, erythema, melanin, elasticity, skin smoothness (Courage-Khazaka) and the volume and depth of nasolabial fold (Primos) were evaluated.

RESULTS

Results *IN VITRO*

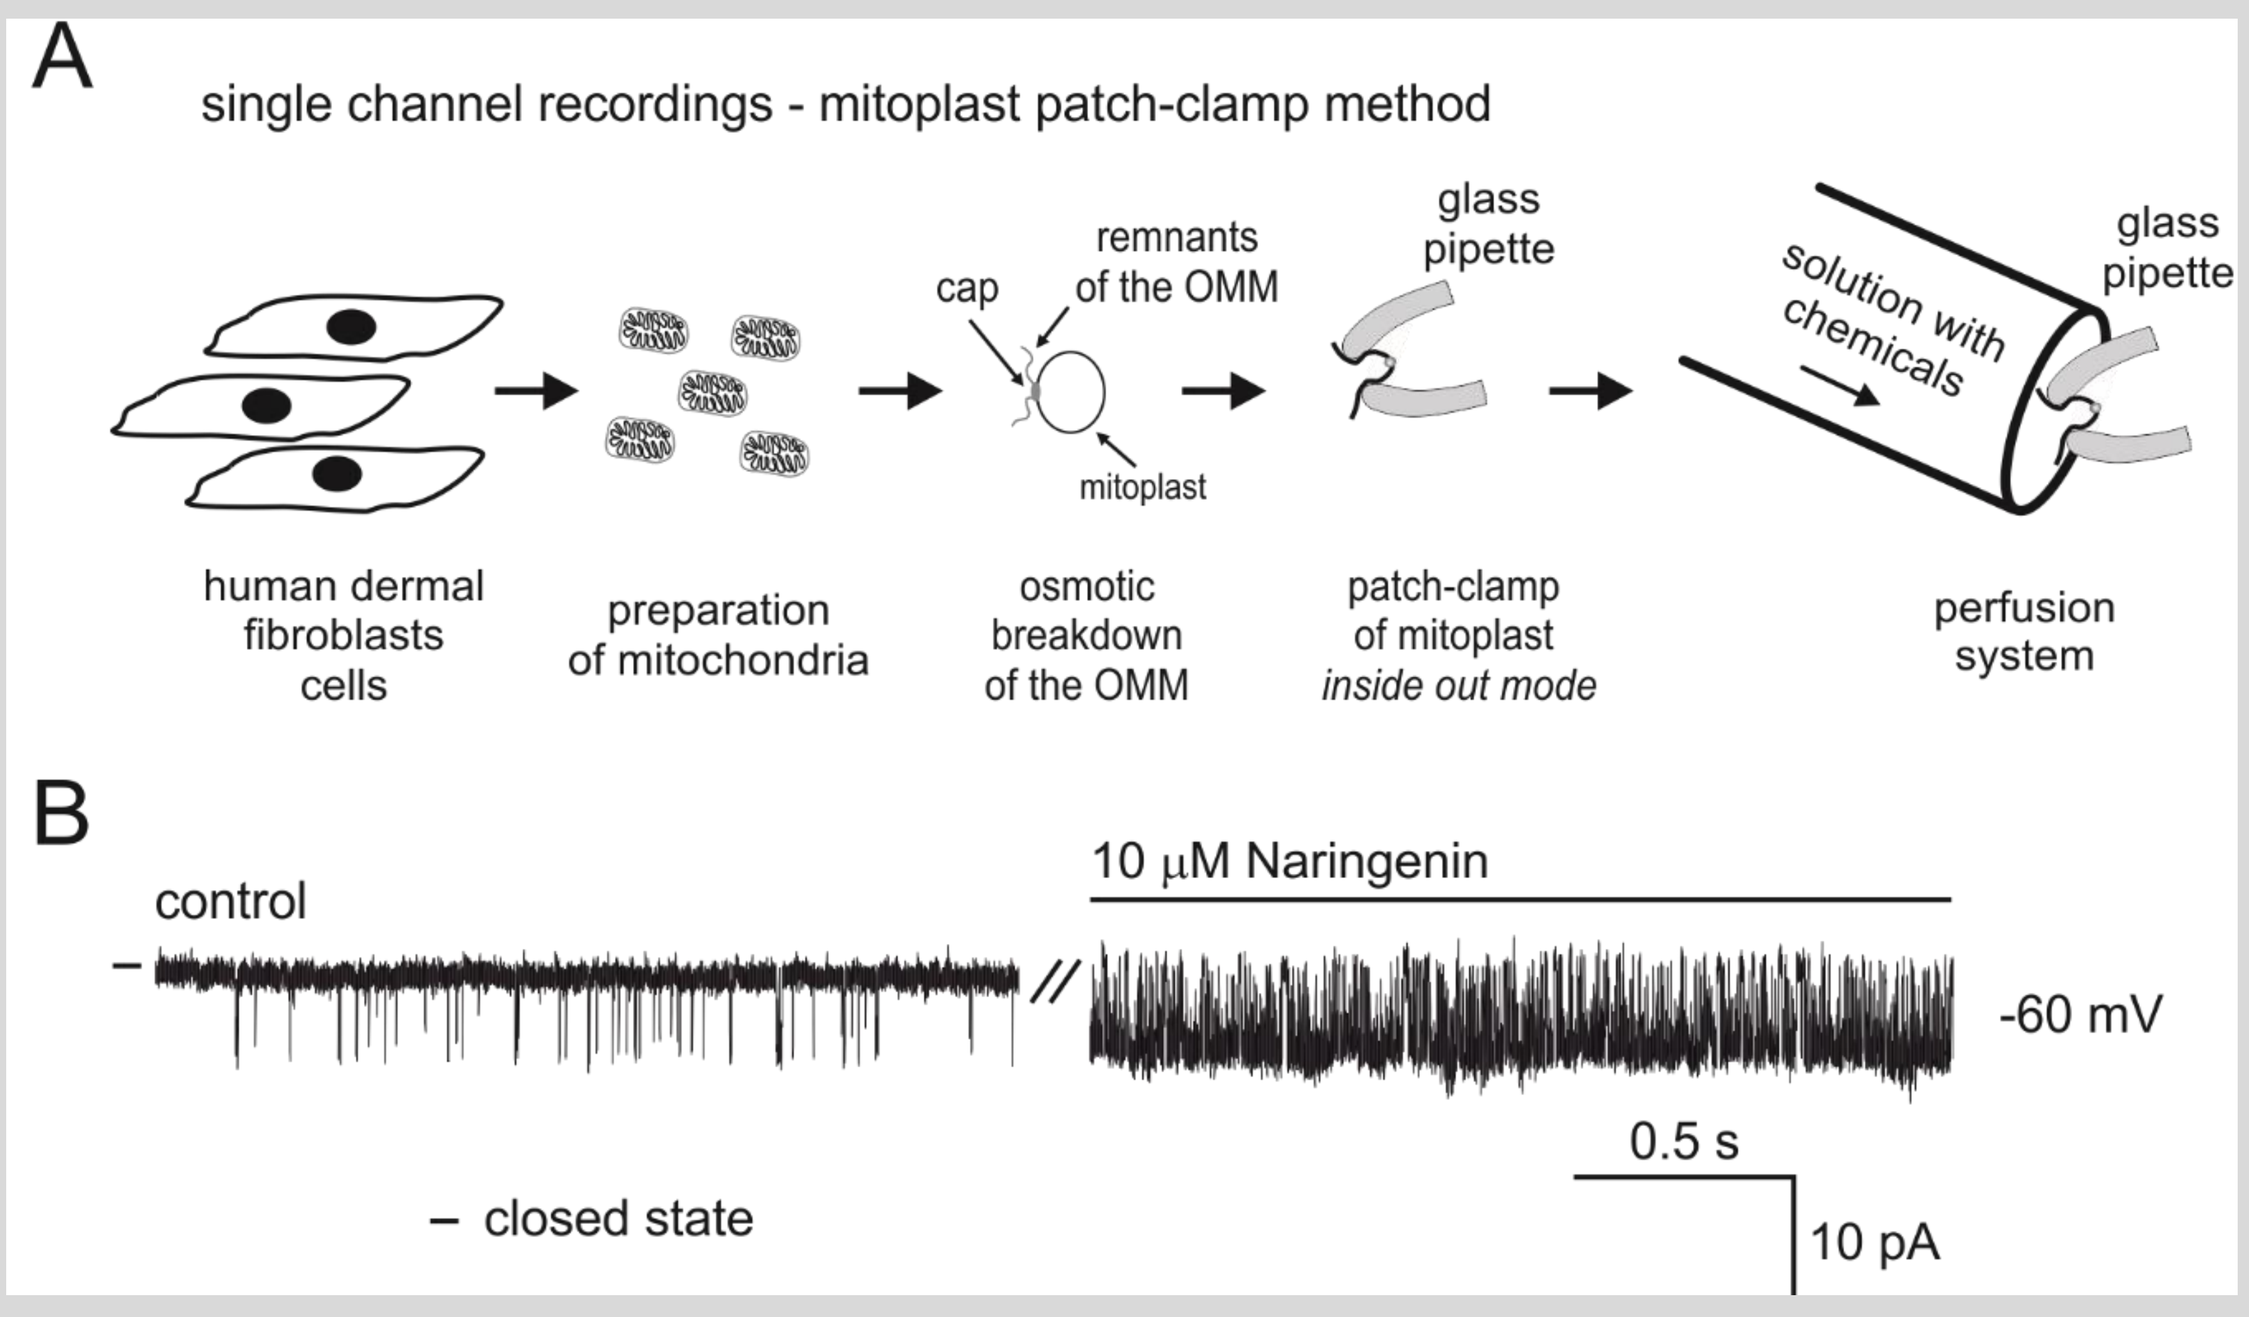
A
single channel recordings - mitoplast patch-clamp method
human dermal fibroblasts cells
preparation of mitochondria
cap
remnants of the OMM
mitoplast
osmotic breakdown of the OMM
glass pipette
patch-clamp of mitoplast inside out mode
solution with chemicals
glass pipette
perfusion system
B
control
10 μM Naringenin
-60 mV
0.5 s
10 pA
– closed state

Figure 1. Influence of naringenin on the activity of the mitoKATP channel from the mitochondria of primary human dermal fibroblasts. (A) A schematic representation of the mitochondria and mitoplast preparation from primary human dermal fibroblasts and the patch-clamp experiment. (B) Representative single-channel current-time recordings of the mitoKATP channel in a symmetric 150/150 mmol/L KCl isotonic solution at −60 mV after 10 μmol/L naringenin. The effect of mitoKATP channel activation by naringenin was observed.

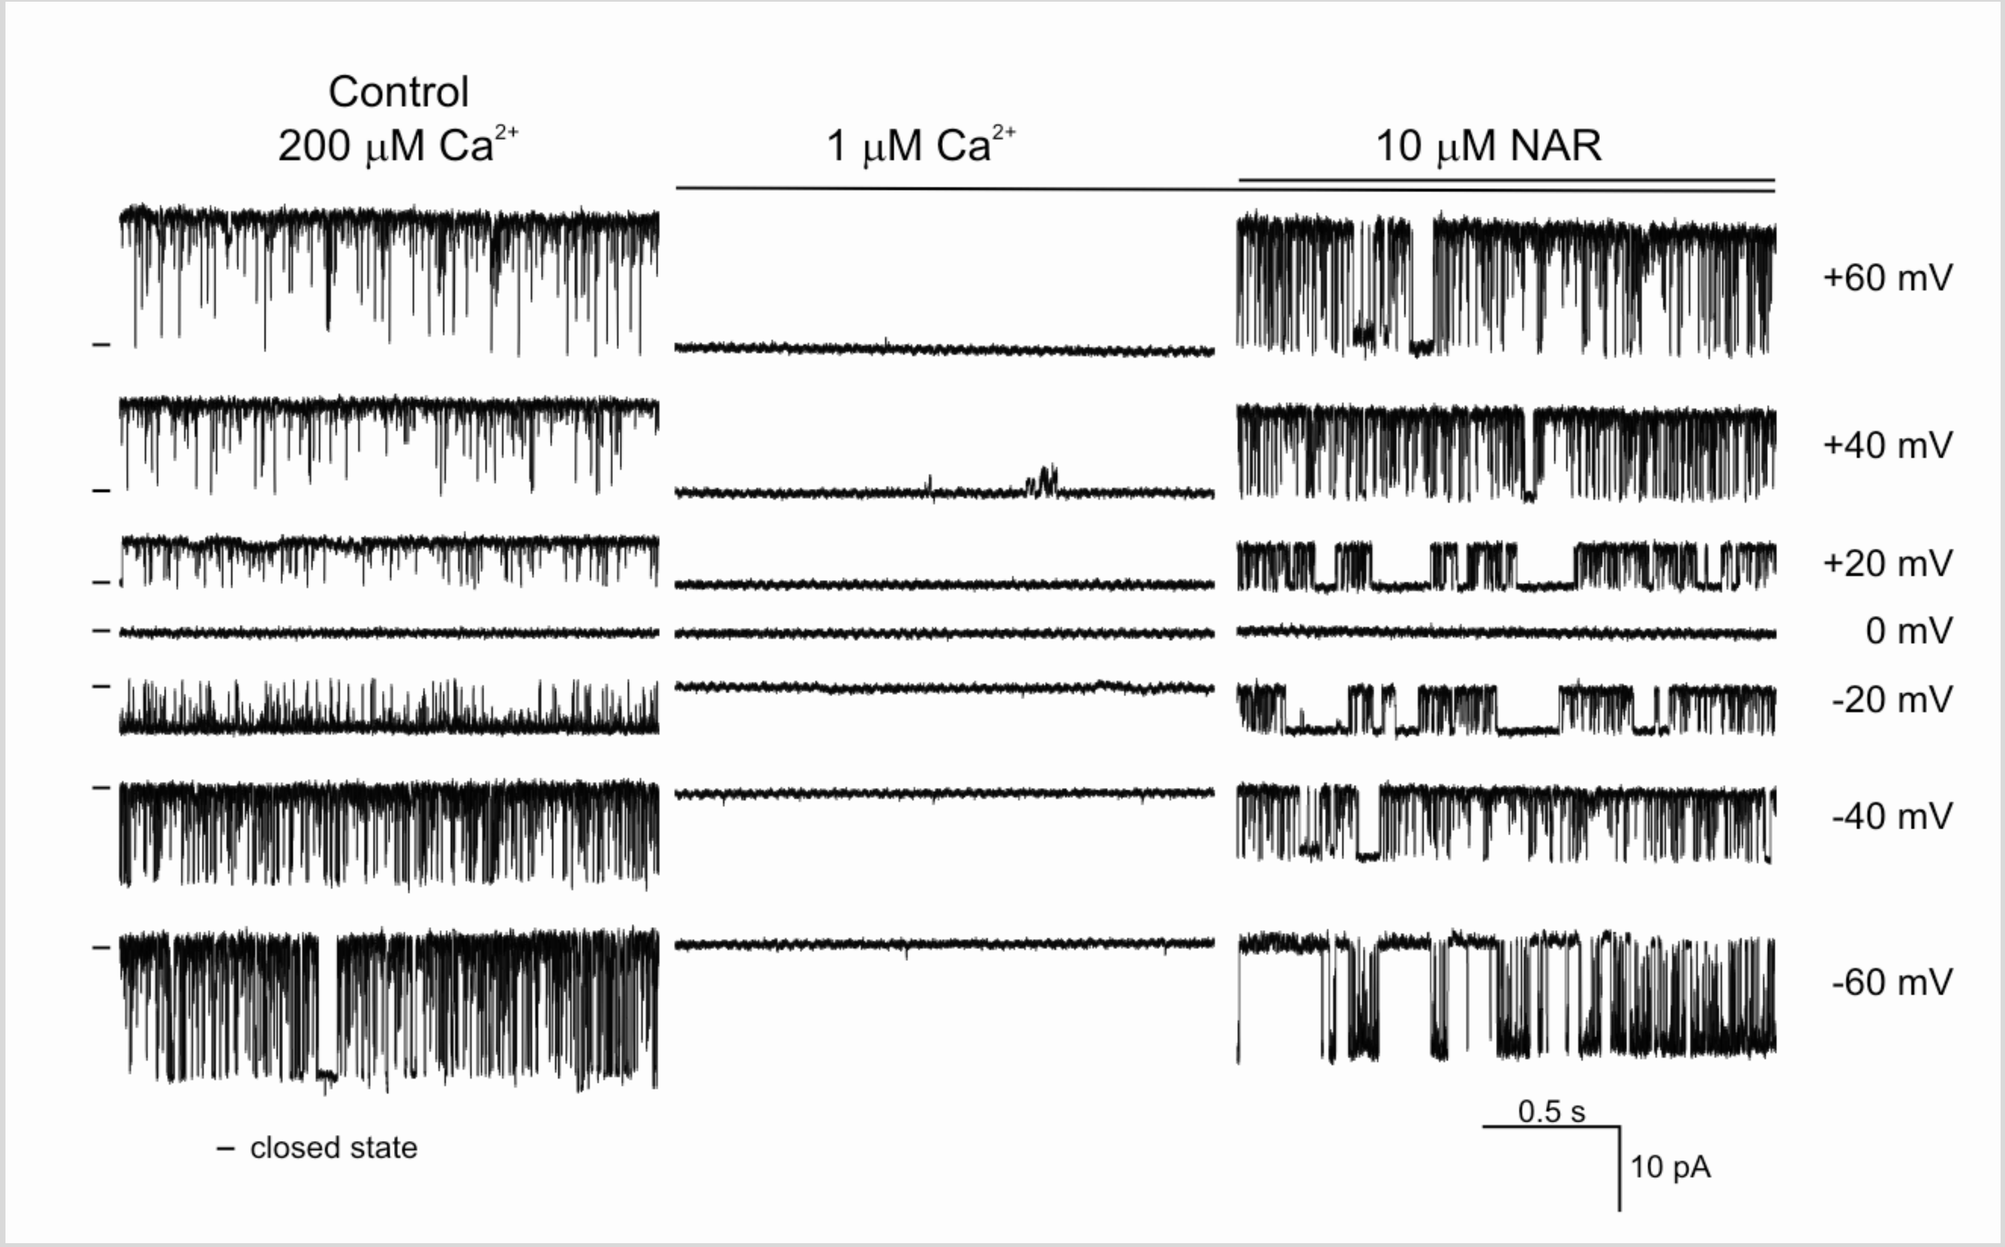
Control
200 μM Ca2+
1 μM Ca2+
10 μM NAR
+60 mV
+40 mV
+20 mV
0 mV
-20 mV
-40 mV
-60 mV
– closed state
0.5 s
10 pA

Figure 2. Effect of naringenin on the activity of the mitoBK_{Ca} channel present in the inner mitochondrial membrane of primary human dermal fibroblasts. Representative traces of single-channel recordings of the mitoBK_{Ca} channel at different voltages in the rage of −60 to +60 mV performed in a symmetric 150/150 mmol/L KCl control solution (200 μmol/L Ca^{2+}), low-calcium solution (1 μmol/L Ca^{2+}) or low-calcium solution containing 10 μmol/L naringenin.

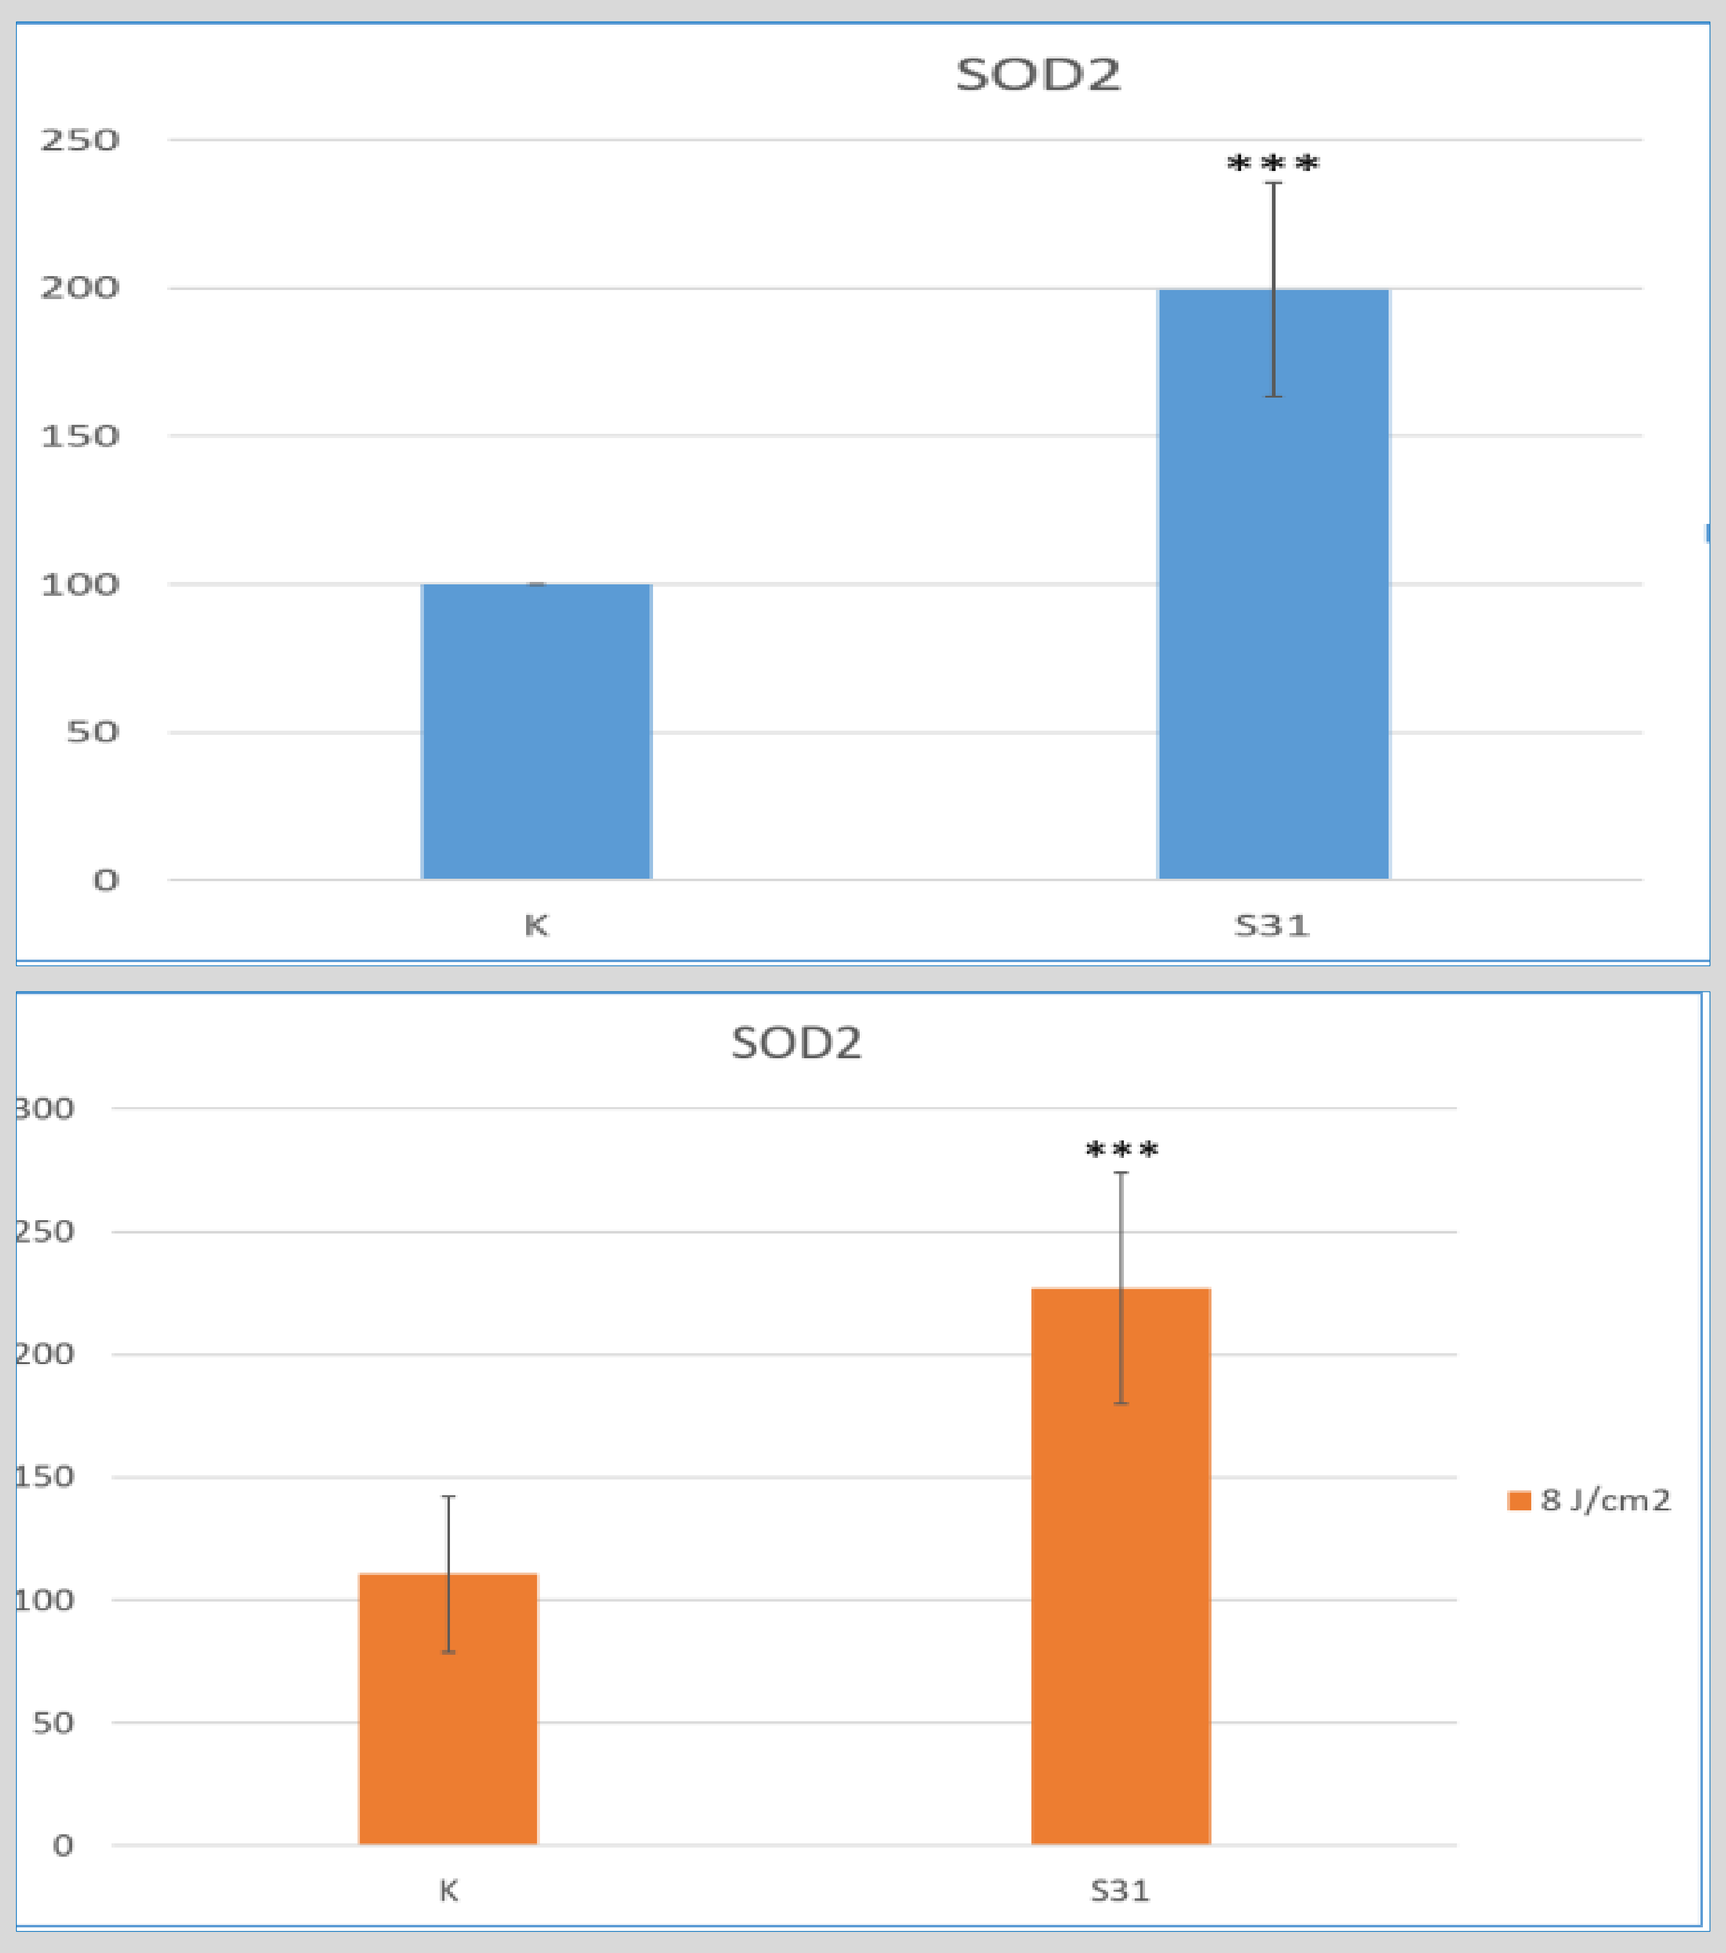
SOD2
K
S31

SOD2
8 J/cm2
K
S31

Figure 3. SOD2 levels increased following incubation with naringenin in both untreated and UVA-irradiated cells by 99% and 127% respectively. K - control, S31 – naringenin.

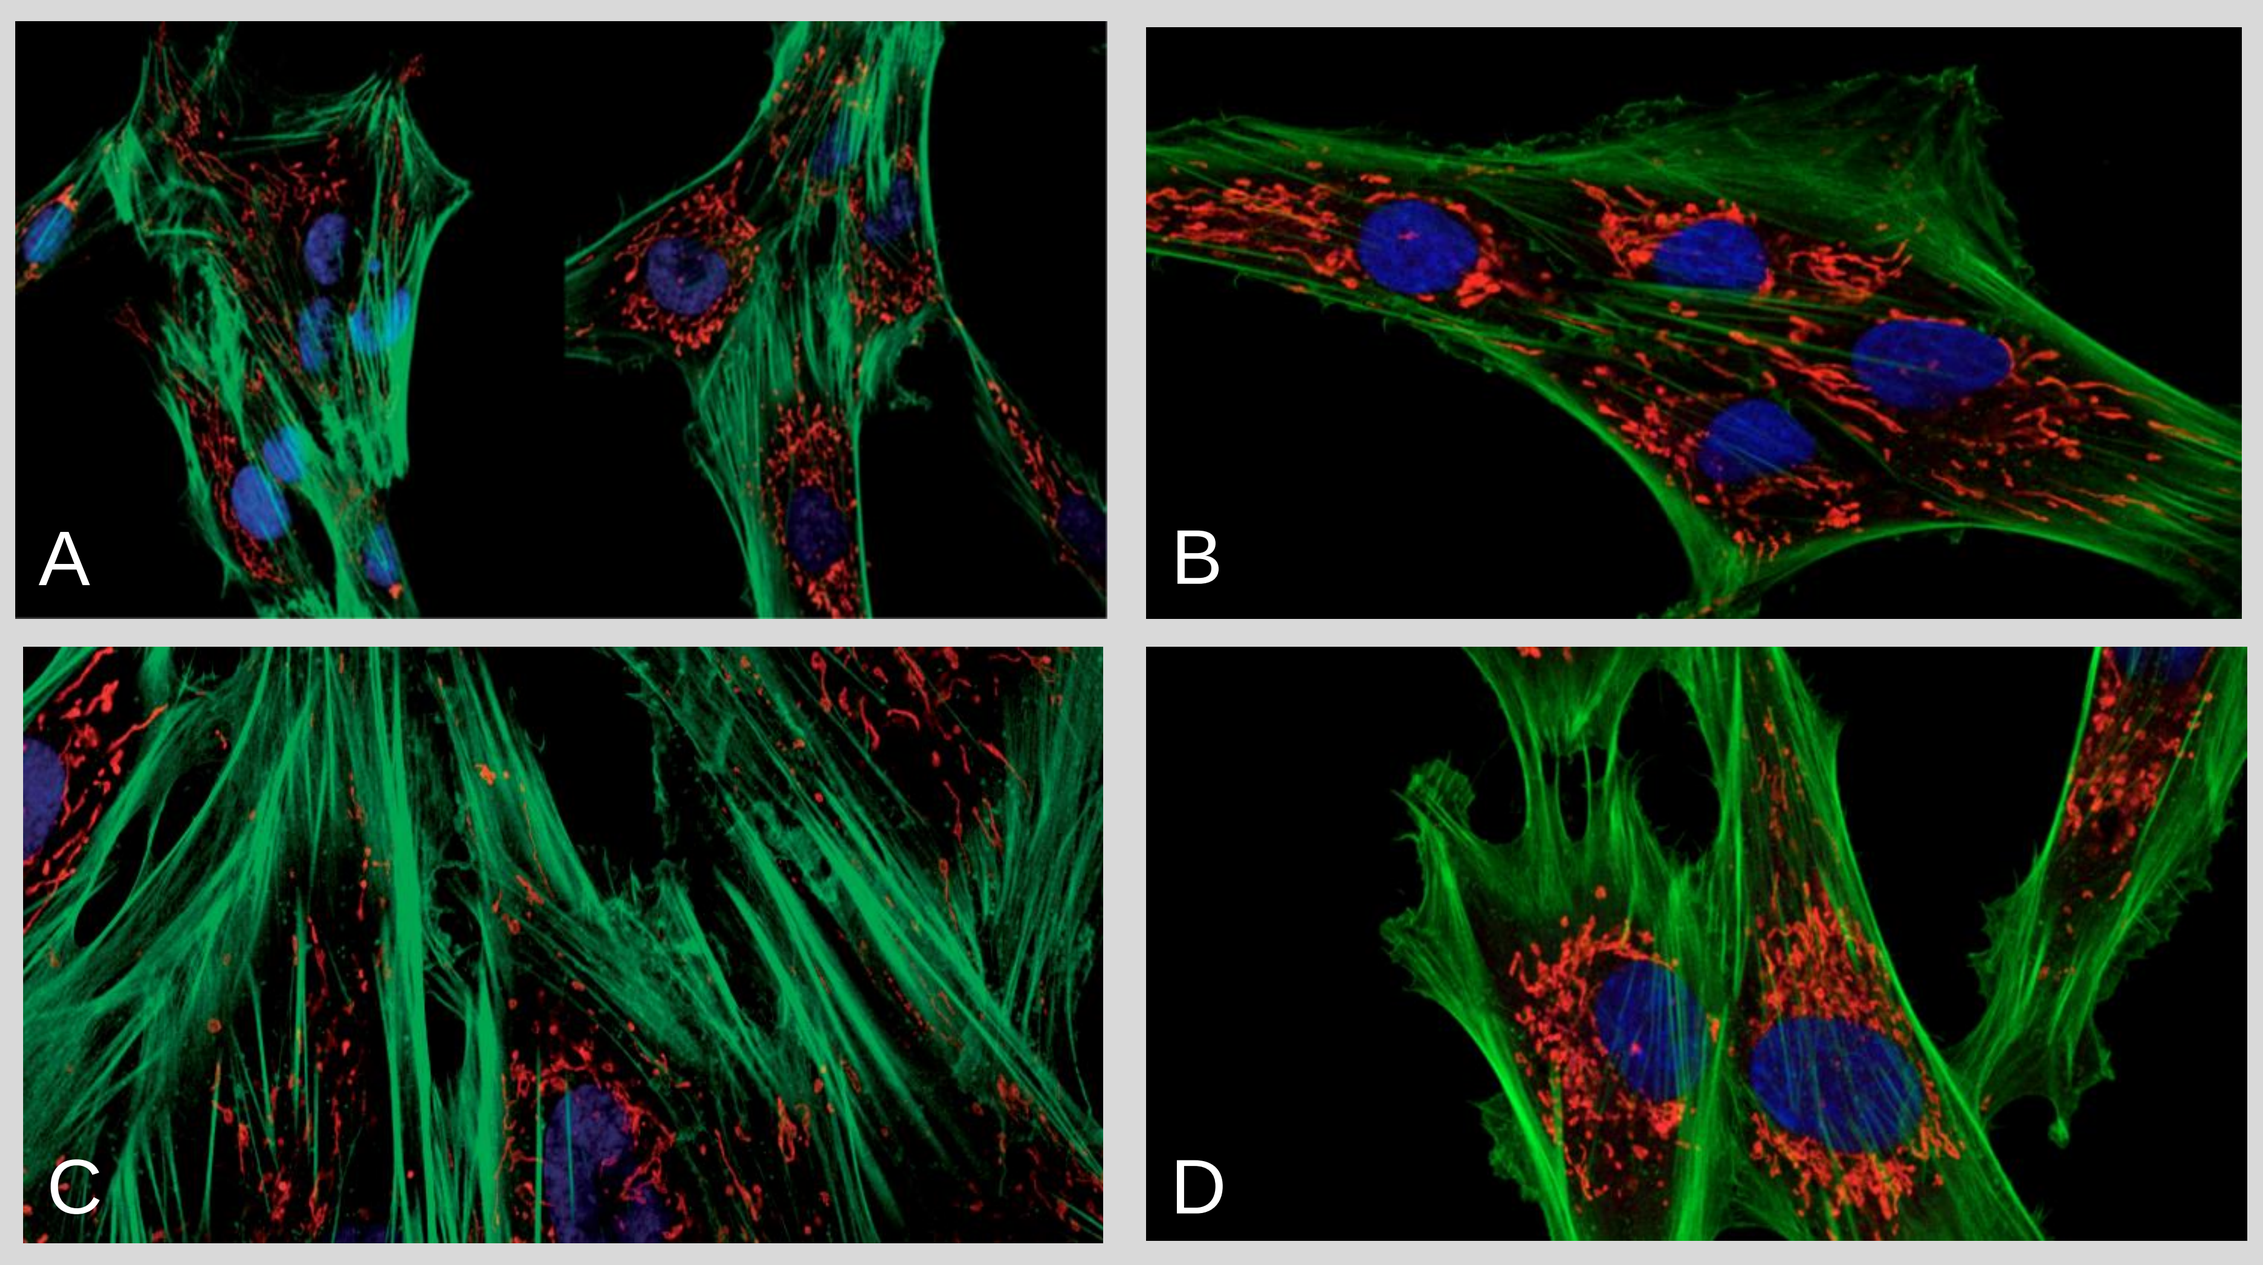
A
B
C
D

Figure 4. Morphology of fibroblasts in confocal microscopy: A – control, B – after naringenin treatement, C – after 8mJ/cm^2 UVA irradiation, D – after 8 mJ/cm^2 UVA irradiation and naringenin treatement. In UVA irradiated cells (C) mitochondria are fragmented and located closer to the nucleus. Mitochondrial network of naringenin stimulated cells after UVA radiation (D) remained identical to the untreated control (A).

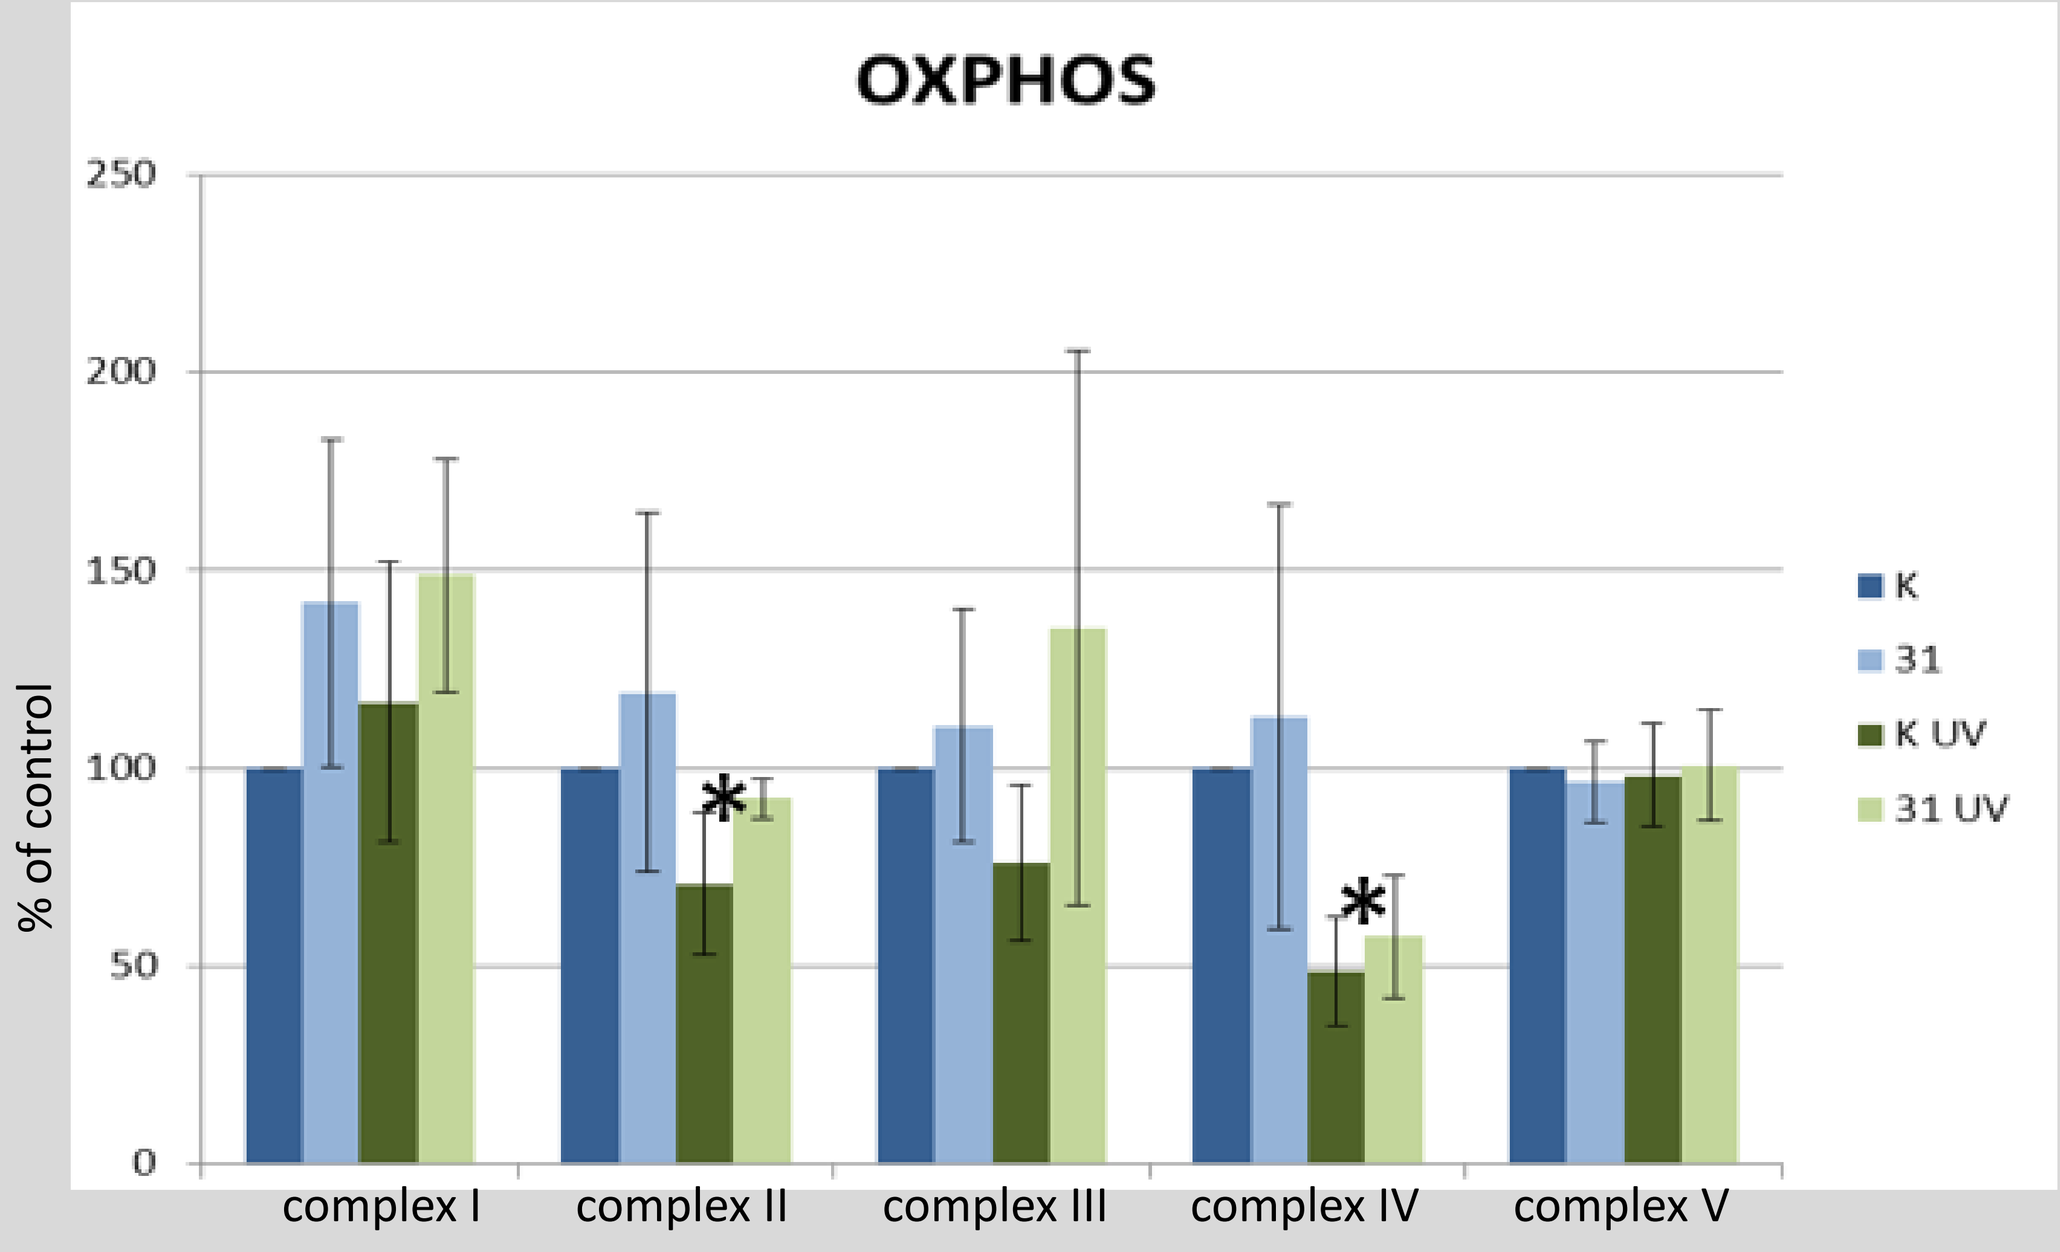
OXPHOS
% of control
complex I
complex II
complex III
complex IV
complex V
K
31
K UV
31 UV

Figure 5. Influence of naringenin treatement and UVA irradiation on the level of respiratory chain subunits (I-IV) and mitochondrial ATPase (V). In the presence of naringenin the level of respiratory chain subunits (I-IV) after UVA irradiation was restored. K - control, 31 – naringenin.

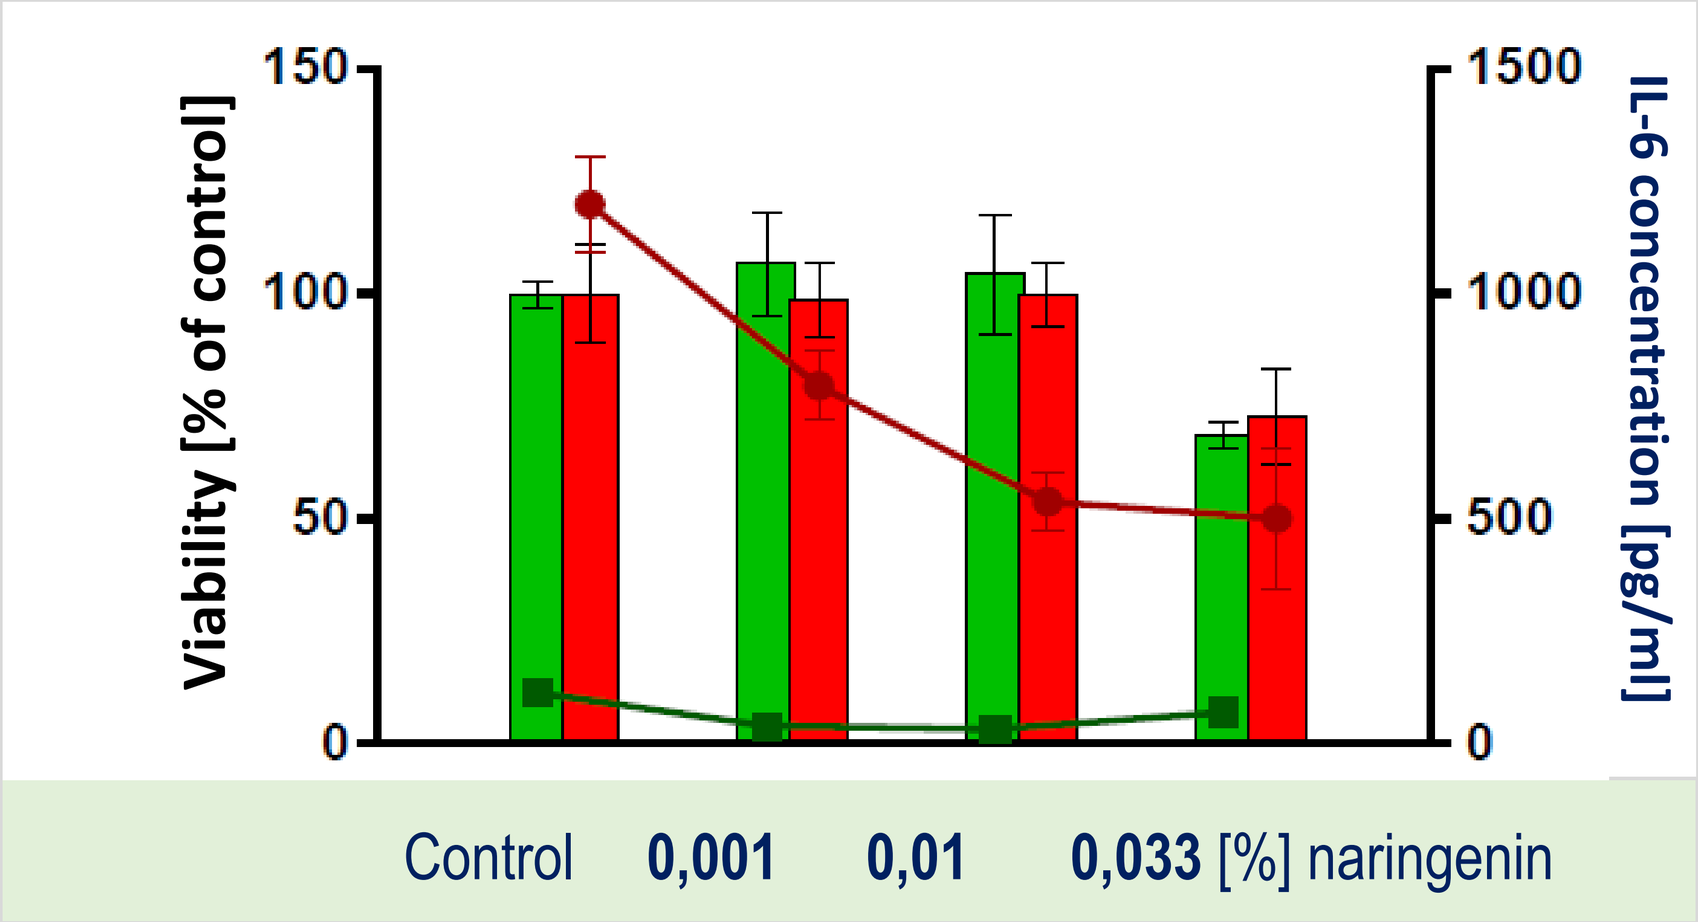
Viability [% of control]
IL-6 concentration [pg/ml]
Control 0,001 0,01 0,033 [%] naringenin

Figure 6. Viability (MTS) and IL-6 levels in HaCaT cells – with or w/o UVB radiation (30 mJ/cm^2). In the concentrations of 0,001% to 0,033% and under UVB-irradiation naringenin inhibited IL-6 production.

Results *IN VIVO*

Skin parameter	Baseline	Average improvement after 4 weeks	Actual improvement after 4 weeks
Scaliness (Sesc) n=19	100%	66%	Reduction of scaliness by 45% in 74% of the study subjects
Skin smoothness (Sesm) n=19		113%	Increase in skin smoothness by 48% in 63% of the study subjects
Volume n=19		76%	Reduction in depth, number and volume of wrinkles by 51% in 58% of the study subjects
Melanin n=22		91%	Skin lightening by 11% in 86% of the study subjects
Erythema n=21		89%	Decrease of erythema by 15% in 76% of the study subjects

Table 1. Change in selected skin parameters (Visioscan, Cutometer-MPA-5) after 4 weeks of treatment with emulsion no. 5552 containing naringenin. The reductions in volume of wrinkles, scaliness, melanin and erythema were observed.

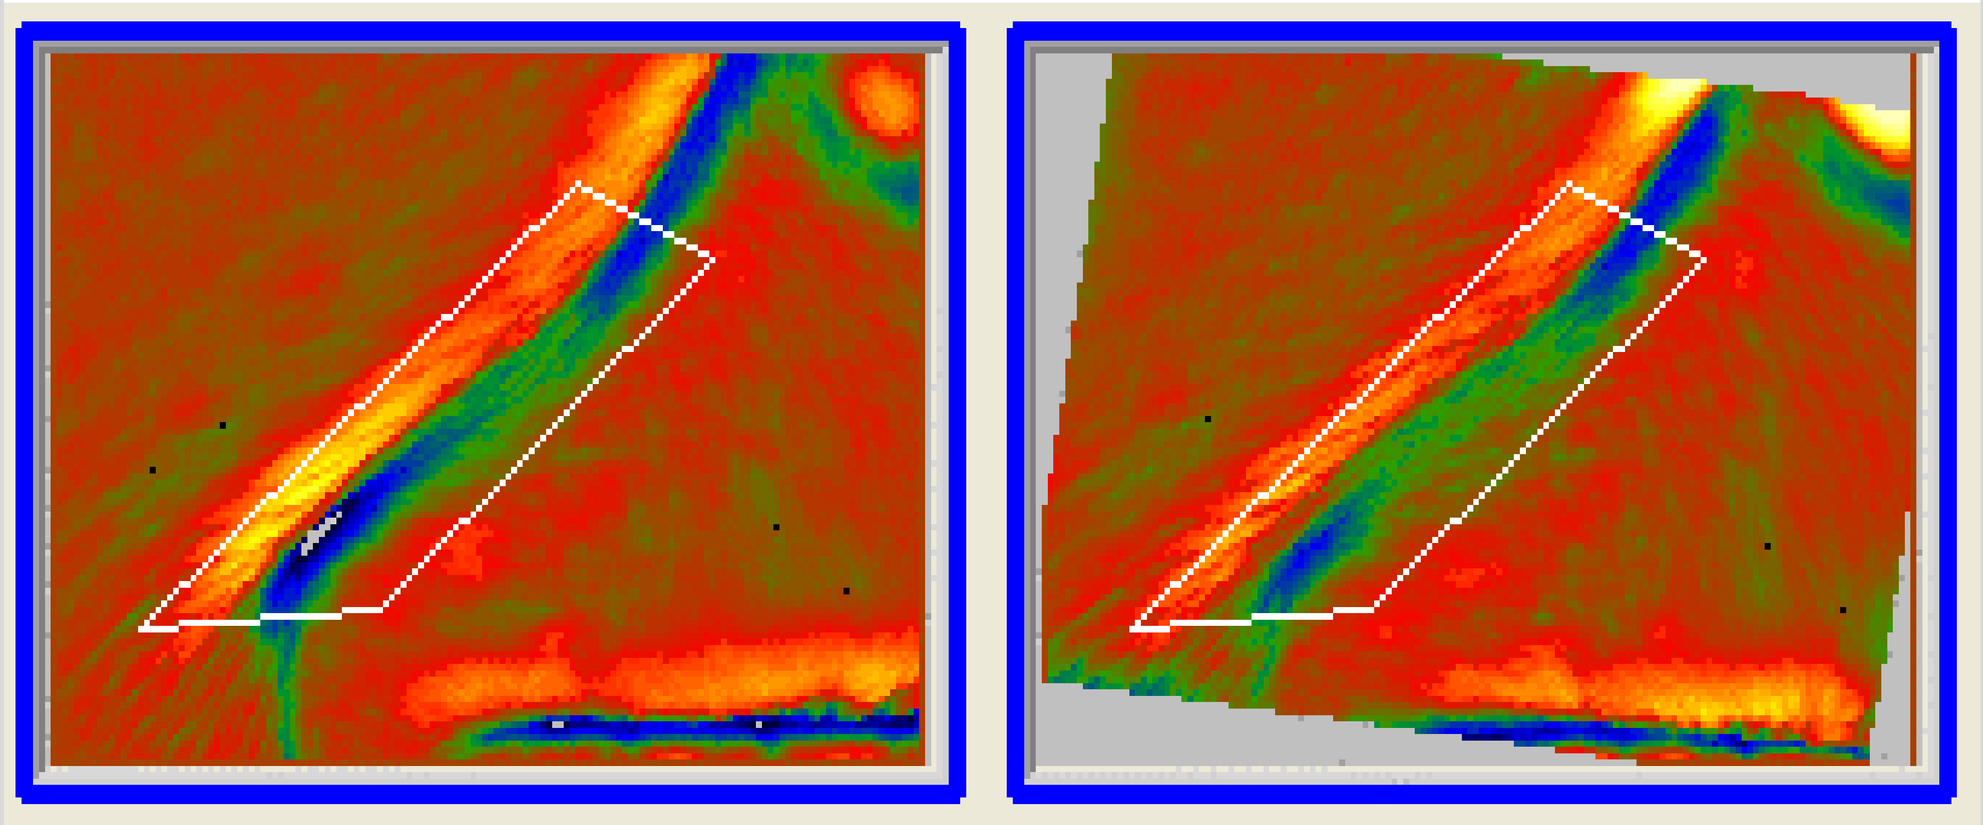

Figure 7. Instrumental evaluation of nasolabial fold volume (Primos) after 4 weeks of using emulsion 5552. The picture shows 64 year old volunteer nasolabial fold before (V=42,22239 mm^3) and after the treatment (V=32,69516 mm^3) revealing 23% (9,52723 mm^3) wrinkle volume decrease.

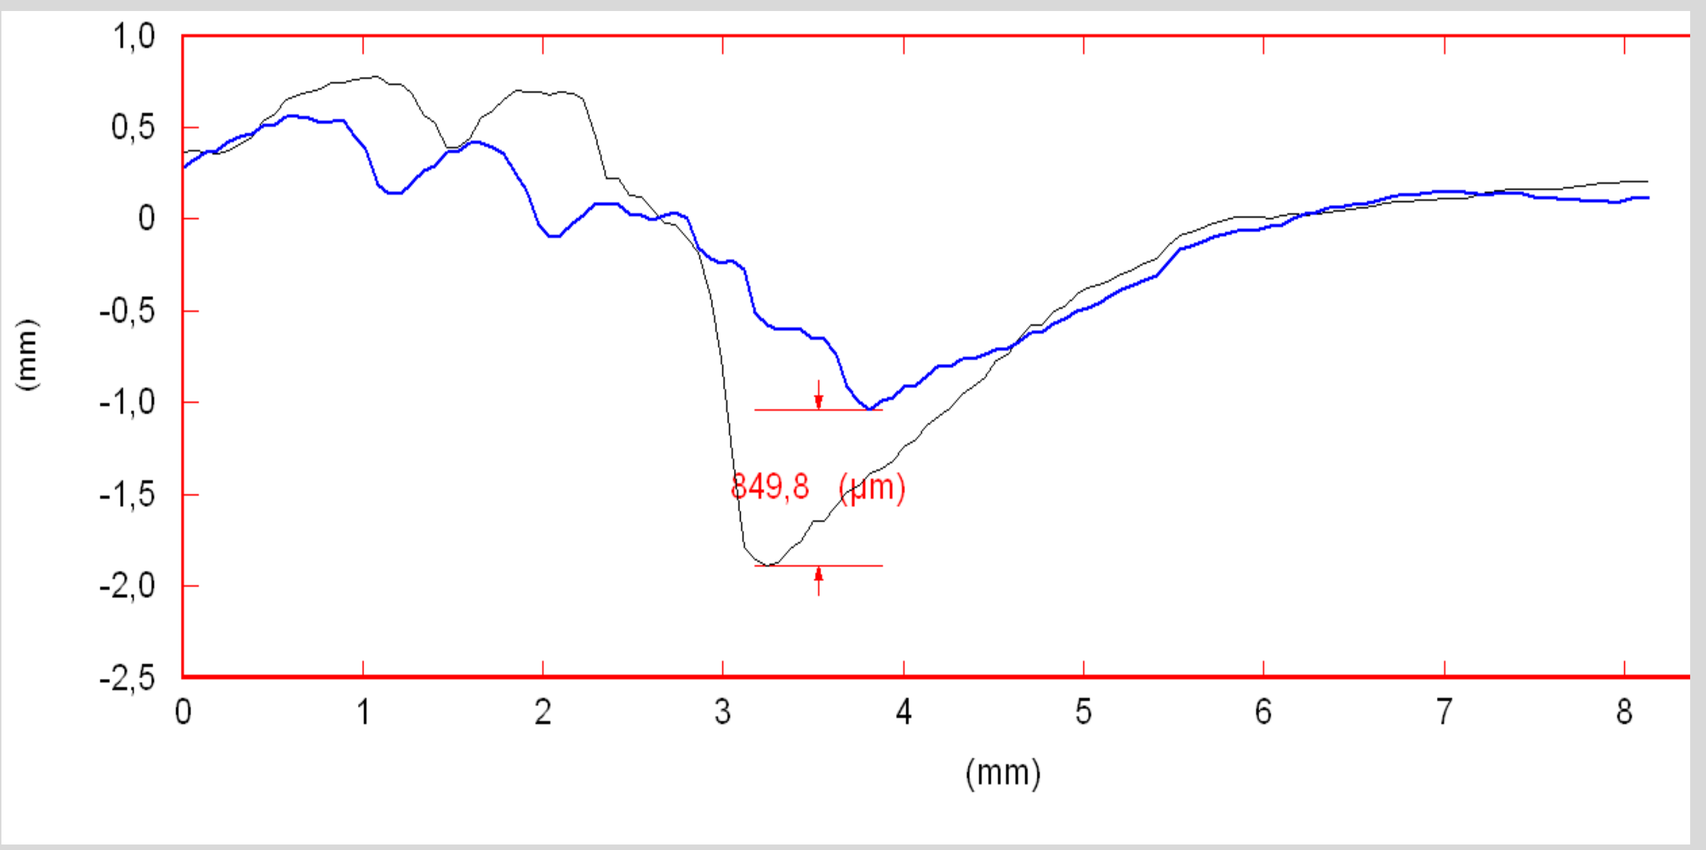
(mm)
849,8 (μm)
(mm)

Figure 8. Reduction of nasolabial fold depth (by 849,8 μm, Primos) in 64 year old volunteer after 4 weeks of using emulsion 5552 containing naringenin (grey line- D0, blue line- D28).

CONCLUSION

The outcome of the study suggests that naringenin is a high potential topical ingredient against mitochondrial ageing through mitochondrial potassium channel regulation. Furthermore, we demonstrated its ability to target Interleukin-6 inflammatory pathways and to reduce the damage caused by excessive reactive oxygen species.

This work was supported by National Centre for Research and Development (NCBR) in the frame of Applied Research Programme (Contract No PBS1/B8/1/2012)