49th Annual ESDR Meeting 2019, Bordeaux, France

Dr Irena Eris
COSMETIC LABORATORIES

Wide spectrum photoprotection in skin with acne, rosacea and after aesthetic treatments

K. Równy, K. Śmigielska, K. Zdroik, B. Ostrowska, M. Dramińska, R. Dębowska, K. Rogiewicz, I. Eris

Dr Irena Eris Cosmetic Laboratories, Piaseczno, Poland

INTRODUCTION

Full spectrum of sun radiation that reaches the atmosphere can be divided according to increasing wavelength into ultraviolet (UV), high energy visible (HEV) and infrared (IR) light. Each one of them can cause skin damage such as sunburn (UVB), photoaging (UVA), inflammatory response and oxidative stress. Therefore photoprotection is a crucial part of the treatment in various inflammatory skin diseases. Protective dermocosmetic formulations should be designed accordingly to a specific skin type.

The aim of the study was to assess safety and efficacy of two formulations with SPF 50+ among patients with dermatoses such as acne vulgaris, rosacea and skin affected by aesthetic treatments.

Both tested formulas provided wide spectrum protection from sun radiation in UV, HEV and IR ranges as well as alleviated symptoms of the respective dermatoses. Emulsion 14004 contained hesperidin (Hesperidine Methyl Chalcone), a flavonoid used in care of rosacea and dilated capillaries as well as hypersensitivity caused by aesthetic treatments (chemical peels, laser therapy). Emulsion 14005 contained piroctone olamine that restricts the growth of harmful microorganisms, prebiotic and pore-diminishing lentil extract to support acne-prone, seborrheic and oily skin.

MATERIALS AND METHODS

SPF value was measured based on ISO-24444:2011 (using Solar Light – UV Solar Simulator 16S-150-001 V4.0.) Emulsion 14904 efficacy against visible and infrared light was measured by spectrophotometric method (Lambda 950) in short cut cuvette (SOWF Journal, 2017, 143 (9): 20-24). Level of protection was determined by the transmission rate of HEV and IR radiation.
The antioxidant activity of the tested emulsion was determined using the 2,2-diphenyl-1- picrylhydrazyl (DPPH) radical scavenging method (SOWF Journal, 1998, 124 (5): 282-284).

In vivo trial of emulsion 14904 included 3 groups of subjects who applied the product for 2 weeks. Group I: 29 subjects aged 25-57 with sensitive skin and capillary problems who needed very high sun protection; participants took a satisfaction survey. Group II: 10 subjects aged 30-65 after aesthetic treatments (mesotherapy, TCA peeling, electrocoagulation, dermapen) – tolerance and efficacy of the product was assessed by a dermatologist; group III: 10 patients with rosacea aged 30-60 undergoing pharmacotherapy (tetracycline, metronidazole, ivermectin) – tolerance and efficacy of the product was assessed by a dermatologist.

Safety and efficacy tests of emulsion 14905 were conducted within two groups of subjects. Group A: 24 subjects aged 18-54 with acne, oily skin, sensitive to sun radiation; all took satisfaction survey, 9 participants underwent biometrical measurements of sebum and porphyrins levels (Sebumeter CourageKhazaka, VISIA Canfield). Group B: 10 patients undergoing pharmacological treatment for acne vulgaris (tetracycline, isotretinoin, clindamycin, benzoyl peroxide). Tolerance and efficacy of the product was assessed by a dermatologist.

RESULTS

Photoprotection and antioxidative potential

emulsion	SPF value	SPF value after 40 min bath
14904	60,2	38,8
14905	61,5	43,1

Figure 1. **SPF values of the two emulsions were 60,2 and 61,5. The protection factor decreased by less than 50% after 40 min bath of the test subjects which indicates that it can be classified as water resistant.**

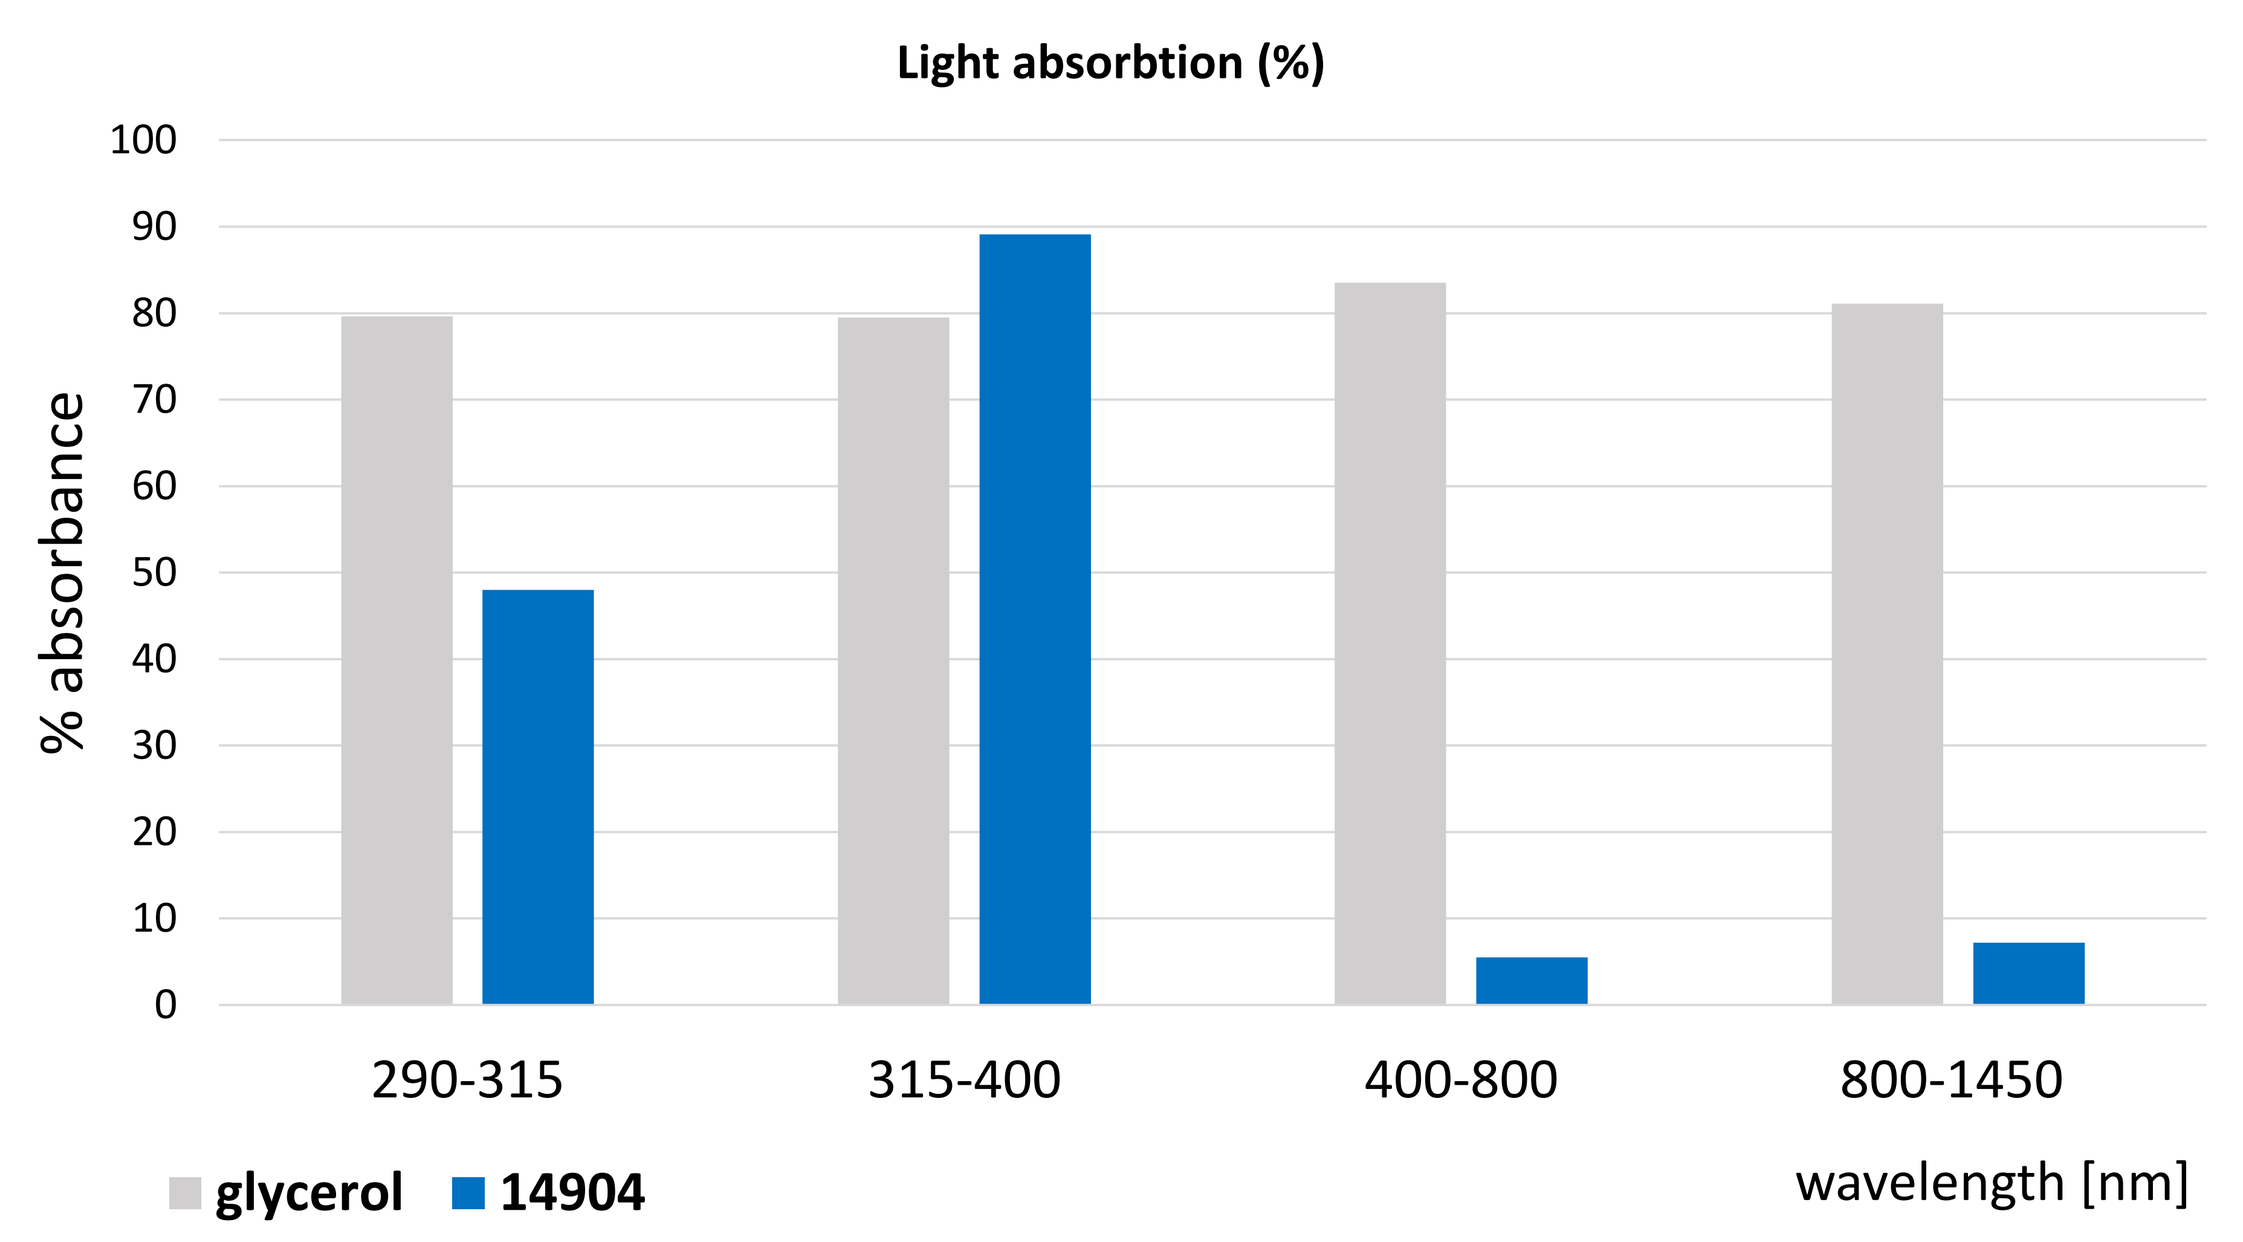
Light absorbtion (%)
% absorbance
100
90
80
70
60
50
40
30
20
10
0
290-315
315-400
400-800
800-1450
glycerol
14904
wavelength [nm]

Wavelenght (nm)	Glycerol [%]	14904 [%]
290-315 (UVB)	79,6	**48**
315-400 (UVA)	79,5	**89,1**
400-800 (HEV)	83,5	**5,5**
800-1450 (NIR)	81,1	**7,2**

Figure 2. Light absorbtion by emulsion14904. Reference sample – glycerol. **Spectrophotometric analysis revealed UVB and UVA protection (48% and 89,1%, respectively) and slight protection against HEV and IR radiation (5,5% and 7,2%, respectively).**

	Antioxidant capacity (mg DPPH/ 1g sample)	RPF (10^{18} DPPH/ 1 g sample)
14904	1,04	1,59

Figure 3. **RPF value (Radical Protection Factor) was 1,59 x 10^{18} DPPH/1g of emulsion and antioxidant capacity was equal to 1,04 mg DPPH/1g of emulsion.**

Safety and efficacy study *in vivo*

Tolerance of tested emulsions as assessed by a dermatologist	
14904	
Group II, n=10 patients after aesthetic treatments	The tested product has very good conditioning properties. It is safe and can be used even by patients with sensitive and capillary skin. Effectively reduces swelling and redness of the skin, as well as itching and burning caused by aesthetic procedures.
Group III, n=10 patients with rosacea	The product is well tolerated by sensitive and hyperactive skin with rosacea. The feeling of transient irritation reported by 1 person resulted in dermatologist opinion from inflammation and skin dryness. 3 women reported difficulties in spreading the cream, its "blunt" consistency, while the other people praised the ease of absorption. The final evaluation of the product is positive.
14905	
Group B, n=10 patients undergoing acne treatments	Very good tolerance in patients undergoing pharmacological treatments. The product meets the requirements of sun protection and care for acne-affected skin.

Figure 4. Opinions of dermatologists who supervised the clinical studies of emulsions 14904 and 14905 in different groups of patients. **Emulsion 14904 was assessed as safe end effective everyday skin care for patients after aesthetic treatments and ones with rosacea. Emulsion 14905 had very good tolerance and was qualified as suitable protective and conditioning product for acne-affected skin.**

Results of patients' subjective assessment	
Emulsion 14904, group I, n=29	**[% subjects]**
minimizes the risk of irritation	97%
minimizes the risk of erythema	97%
minimizes the risk of formation of lasting erythema and telangiectasia	79%
protects the capillaries against hyperreactivity to external factors	86%
strengthens the protective barrier of the epidermis	90%
Emulsion 14905, group A, n=24	**[% subjects]**
reduces the visibility of acne lesions, imperfections	61%
reduces the visibility of inflammatory acne lesions	76%
relieves inflammation	76%
prevents the formation of new acne lesions and imperfections	52%
Provides long lasting mattifying effect	67% (lasts av. 6,7 h)

Figure 5. Selected results of satisfaction surveys after 2 weeks use of emulsions 14904 and 14905 presented as % of subjects who confirmed respective benefits of product use. **Emulsion 14904 was especially well assessed for minimizing risk of irritation and erythema (97% of subjects) and strengthening protective barrier of the epidermis (90%).**
Use of emulsion 14905 was associated with reduced visibility of inflammatory acne lesions and with decrease in inflammation (76% of subjects). It was said to provide long lasting mattifying effect by 76% of subjects with average declared duration of 6,7h.

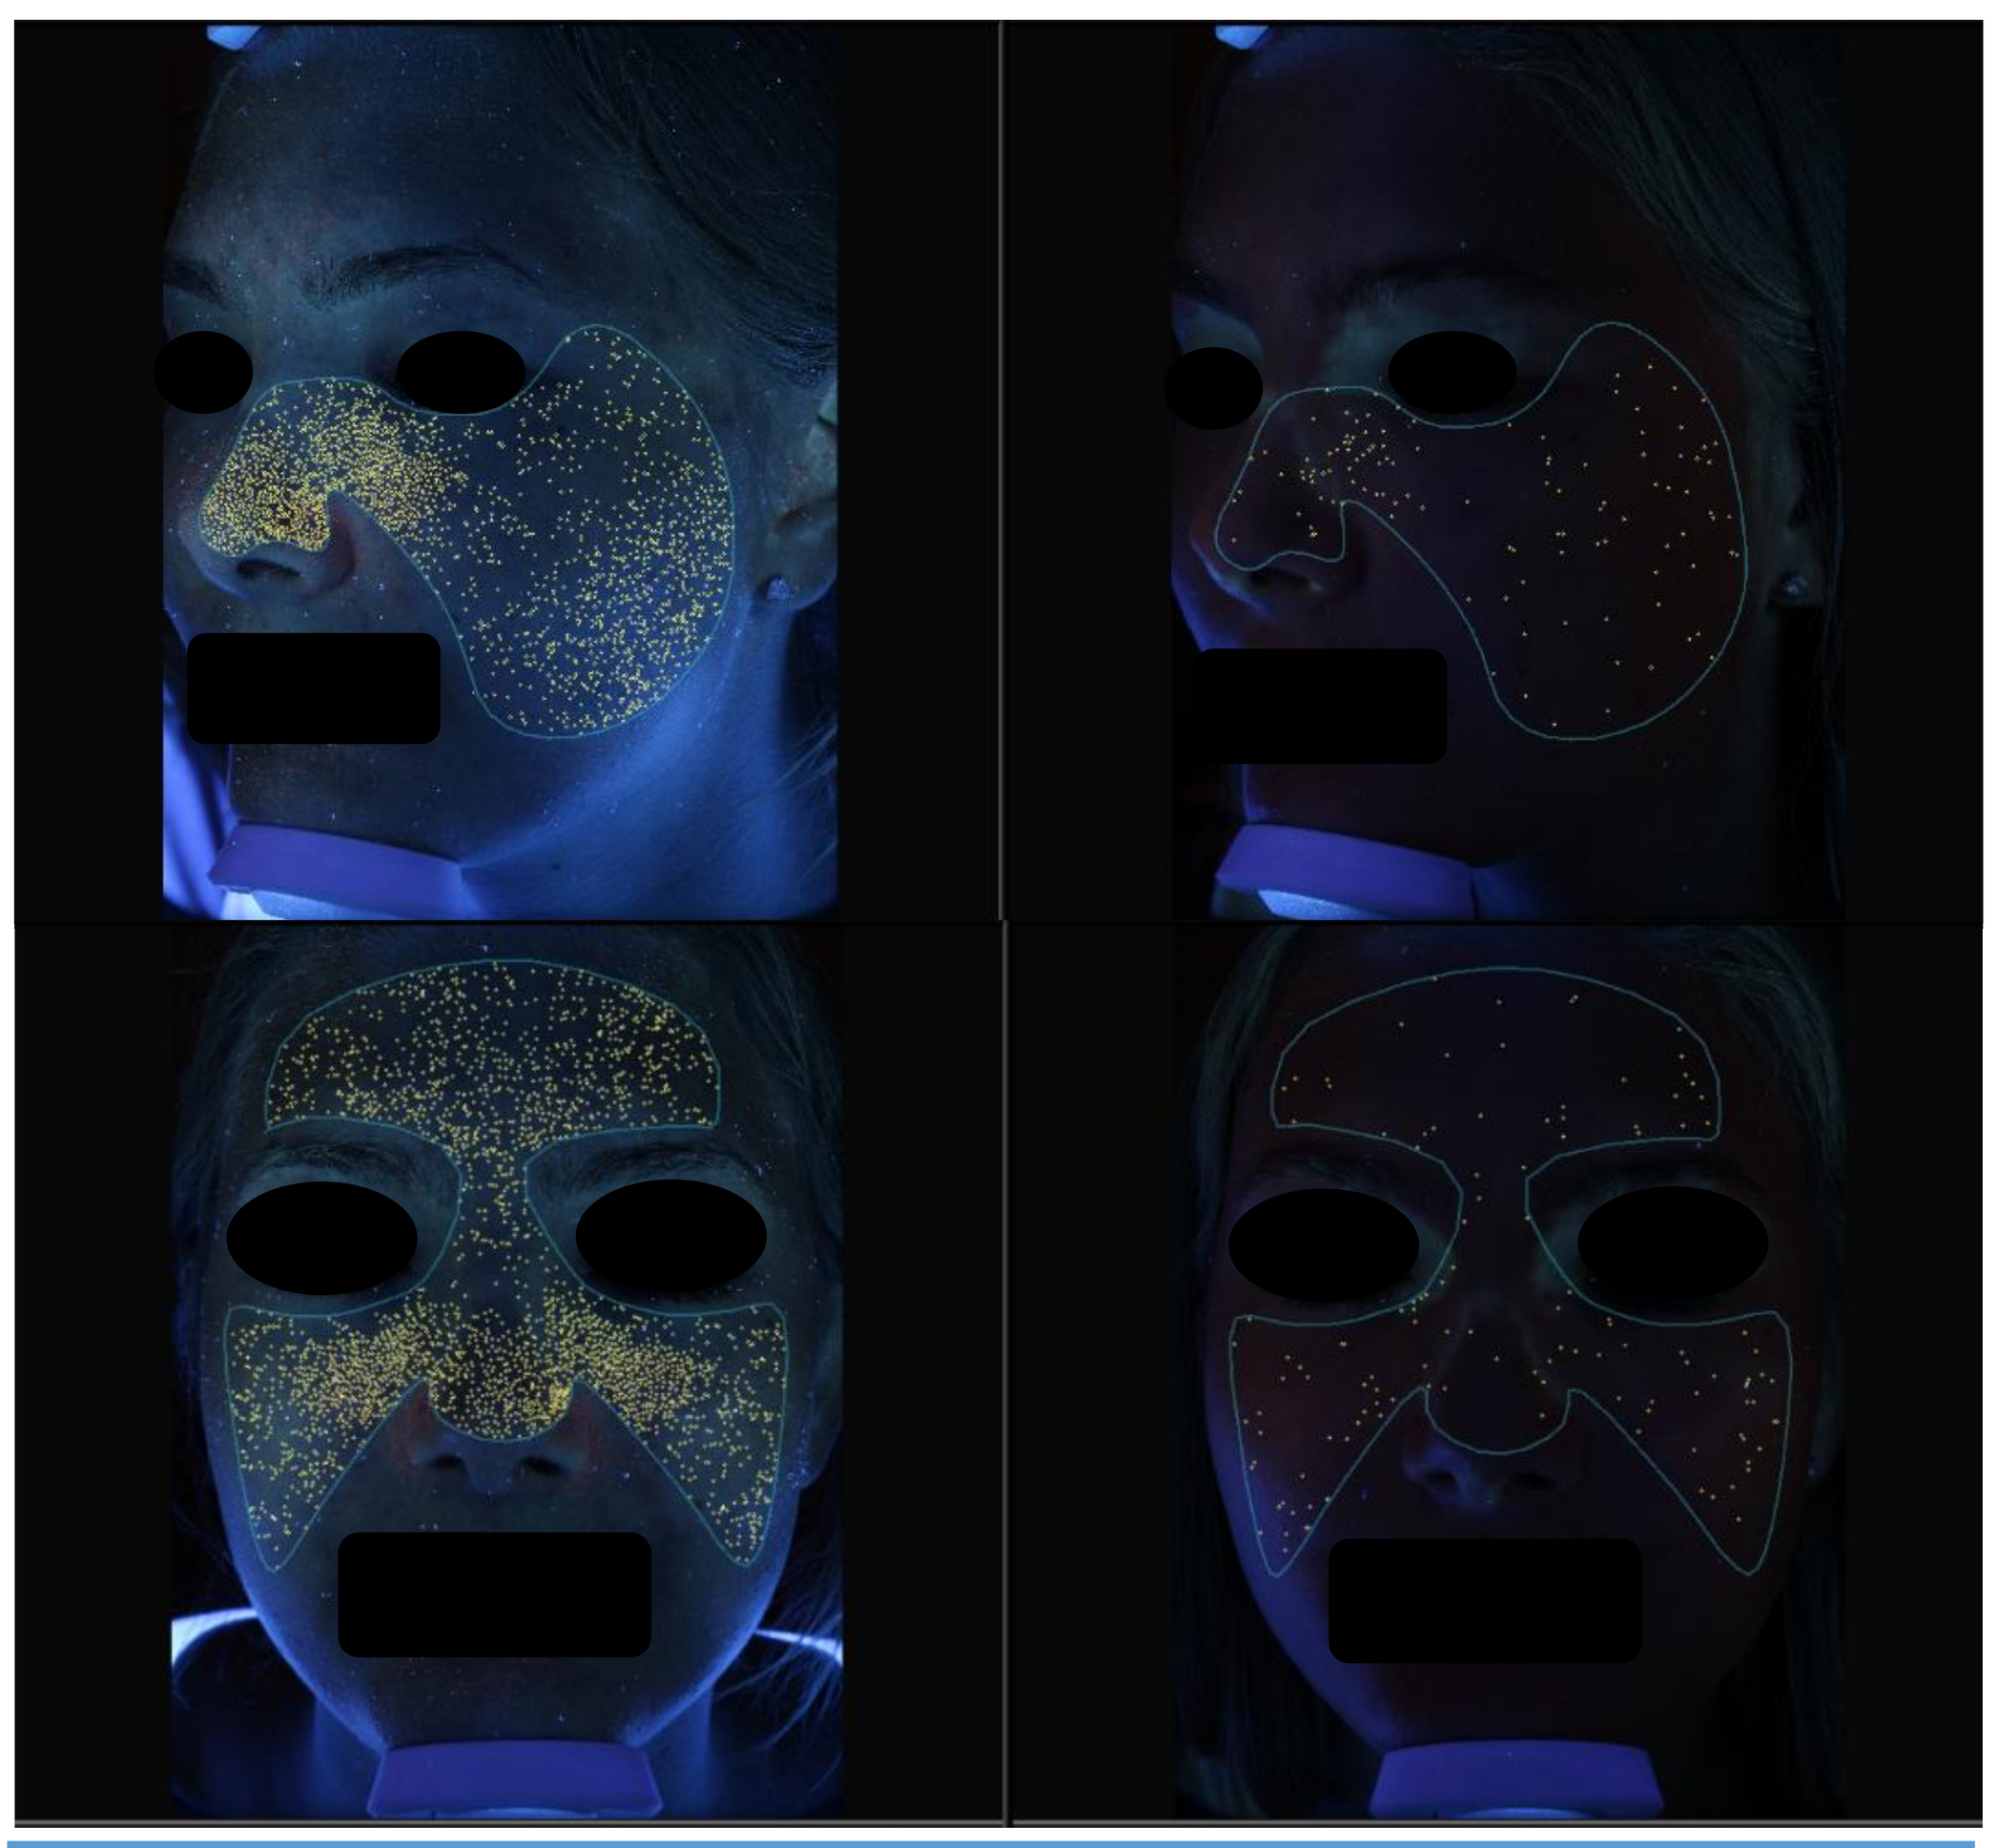

Before and after 2 weeks of using emulsion 14905, group A, n=9	
measured feature	**% change**
porphyrins	-55%
sebum	-19%

Figure 6. Results of instrumental measurements after 2 weeks use of emulsion 14905. The top panel shows **95% decrease in detected porphyrins** in 17 y.o. subject (left profile and *en face*). The table presents results in the whole group – **55% decrease in number of detected porphyrins and 19% decrease in sebum levels**.

CONCLUSION

Presented results confirm the utility and efficacy of dual function products that provide very high protection from sun radiation as well as active ingredients designed for needs of specific skin type. Standardized tests confirmed high SPF value as well as water resistance of both emulsions. In vitro light absorbtion test of emulsion 14904 showed protection from 5,5% HEV and 7,2% IR which is non-negligible in patients with capillary-related issues like rosacea. Along with its high antioxidative potential the emulsion provides high anti-free radical protection.
Emulsion 14905 caused the average of 55% decrease in detected porphyrins and 19% decrease in sebum levels in patients after 2 weeks of application.
Both products were assessed as safe and effective by supervising physicians as well as patients.