53th Annual ESDR Meeting 2024, Lisbon, Portugal

Dr Irena Eris
CENTRE FOR SCIENCE AND RESEARCH

Dermal safety evaluation of two plant polyphenols: ellagic acid and glabridin

Karolina Lendzion, Monika Pasikowska-Piwko, Renata Dębowska, Katarzyna Rogiewicz, Irena Eris

Dr Irena Eris S.A., Department of Research and Innovation, Centre for Science and Research, Piaseczno, Poland

INTRODUCTION

Proper skin sensitization and irritation data of the individual chemicals is essential, especially when dermal contact is intended, like for cosmetics. In some cases, the combination of individual ingredients may also require evaluation in order to reveal possible mixture effects.

The aim of this study was to evaluate the dermal safety of two plant-derived polyphenols tested separately: ellagic acid (EA) and glabridin (GLB) *in vitro* and three cosmetic formulations with a combination of both ingredients (5-10 µg/mL) in human trials.

MATERIALS AND METHODS

The safety of the ingredients was evaluated in accordance with ISO 10993 by performing *in vitro* irritation and sensitization potential on Epiderm skin model. Briefly, 25 mg of powdered substances were applied on tissue inserts (n=3) and incubated for 1 h. After that time, tissues were washed and exposed to 24 h post-treament incubation. Then, culture medium was collected to perform ELISA analysis of interleukin-18 concentration, and tissues were incubated for additional 18 h. Next, tissue viability assay was performed to assess skin irritation potential. Phototoxicity was evaluated using 3T3 NRU assay in Balb/c 3T3 cells.

Patch tests (including skin irritated with 10% lactic acid) of each formulation (day cream 14711, night cream 14712, and eye cream 14713) were tested in a total group of 150 subjects. In addition to this, skin redness (Mexameter) before and after 4 weeks of three formulations usage was performed on additional group of 30 patients. Moreover, the application test was performed in a group of 30 pregnant or breastfeeding females.

RESULTS

In vitro study confirmed that pure ingredients tested separately did not express irritation potential on EpiDerm model (tissue viability for glabridin: 91.6% and ellagic acid: 97.5%; **Fig. 1**) while compared to untreated control tissues. For both EA and GLB, the concentration of IL-18 was significantly lower compared to negative control (**Fig. 2**). Neither EA (**Fig. 3**), nor GLB (**Fig. 4**) exhibited phototoxicity at concentrations from 4.64 to 1000 µg/mL. All safety human studies (including pregnant females) displayed high tolerance of all three cream formulations (**Tables 1** and **2**), as well as reduction in skin redness in objective measurements (**Table 3**).

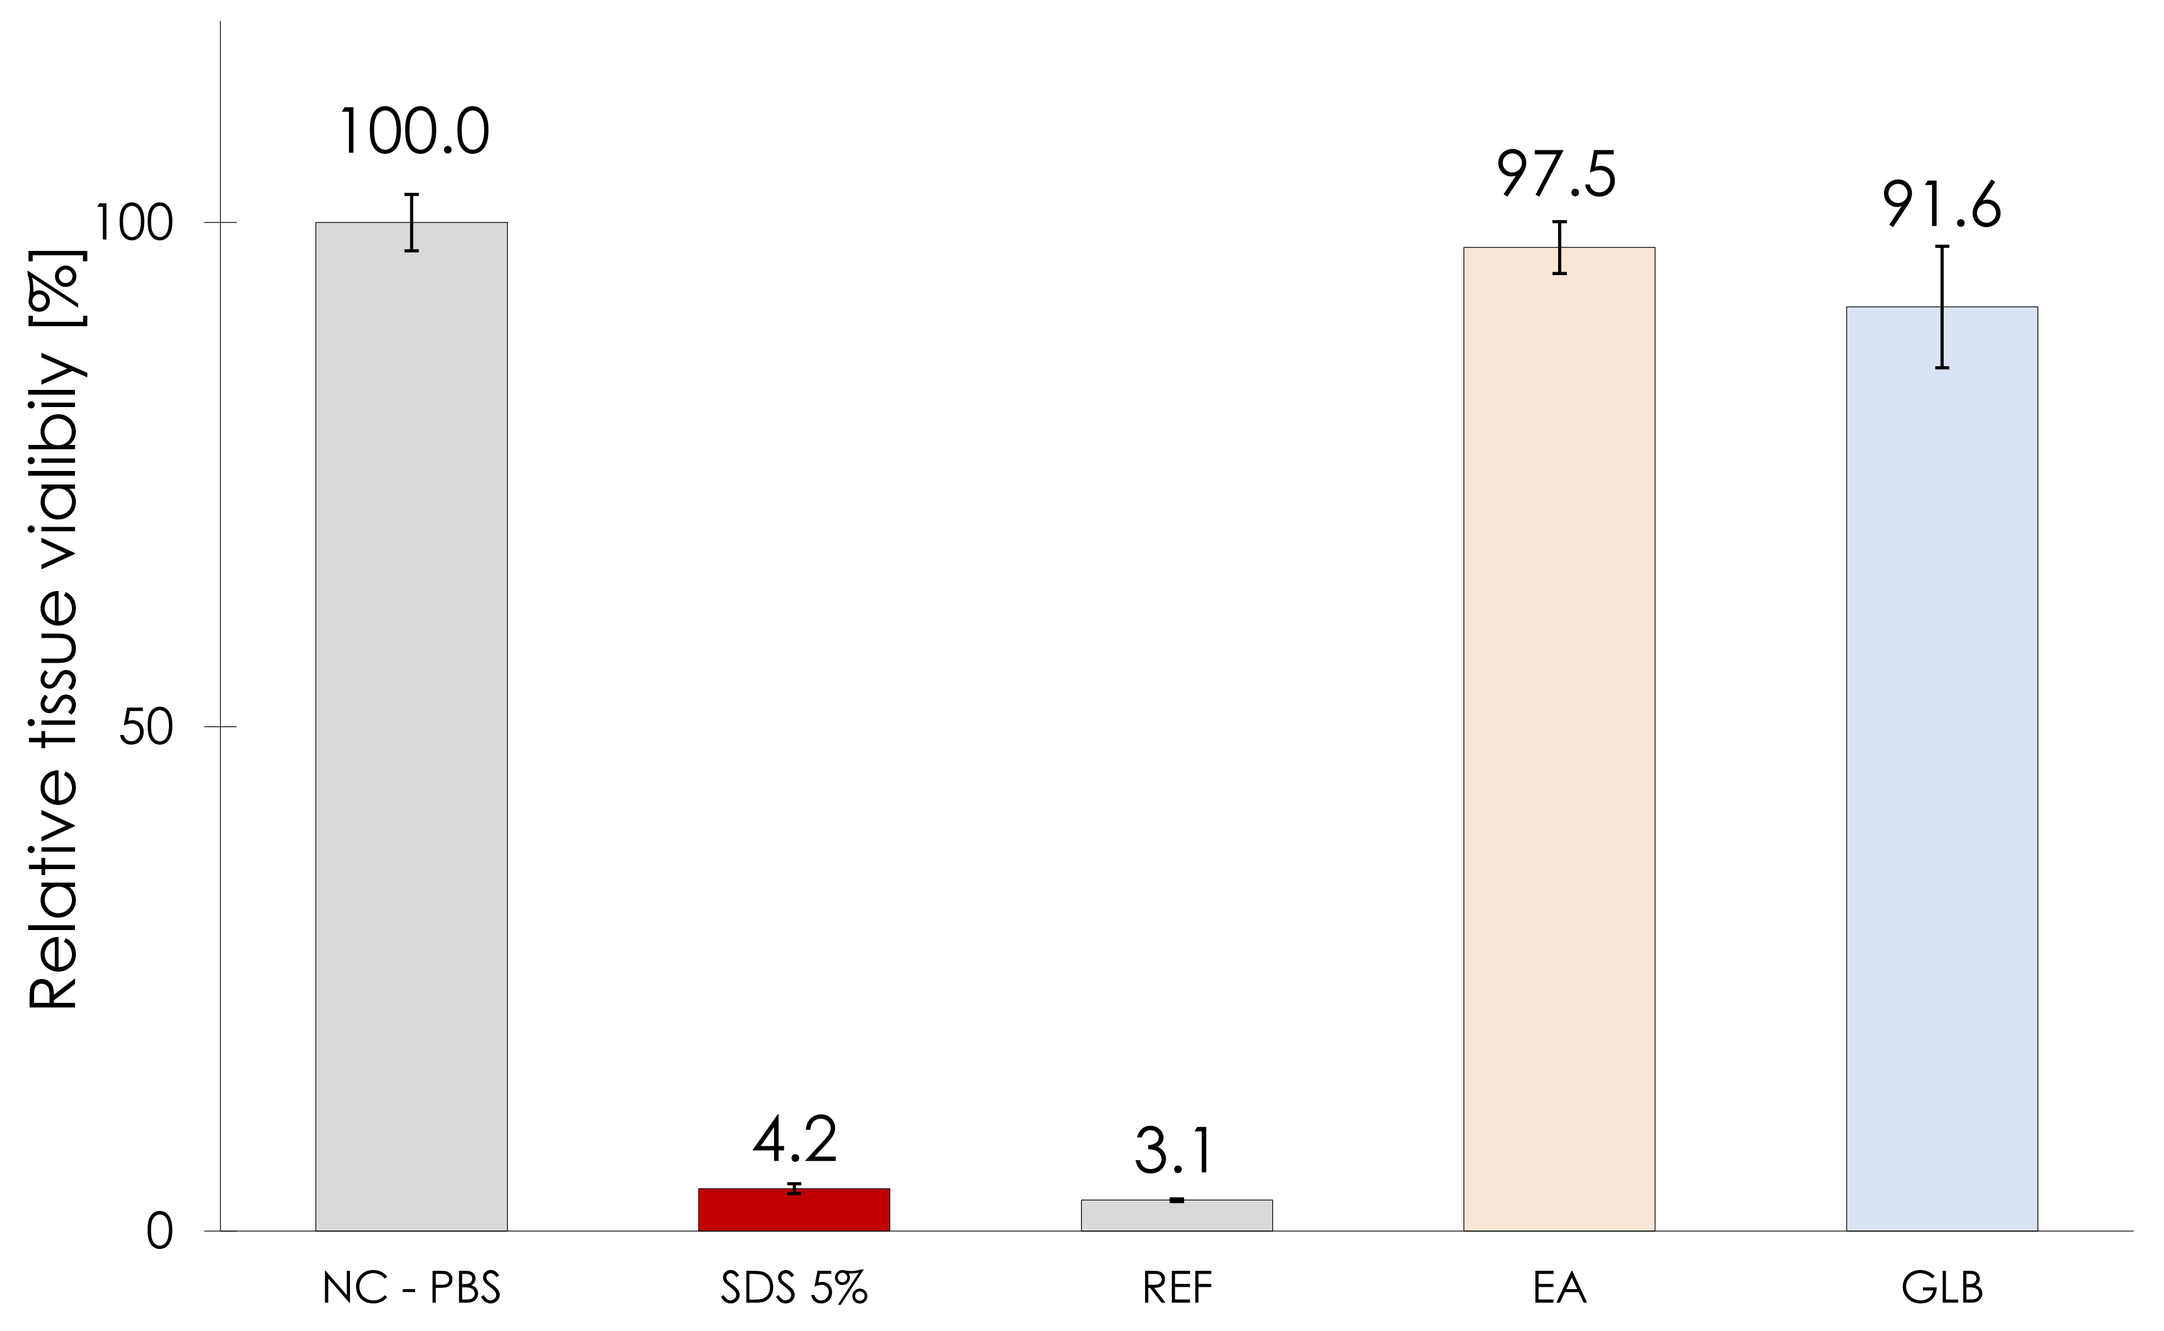
Relative tissue vialibily [%]
100.0
4.2
3.1
97.5
91.6
0
50
100
NC - PBS
SDS 5%
REF
EA
GLB

Figure 1. In vitro skin irritation potential (EpiDerm model) of ellagic acid (EA) and glabridin (GLB). Results are presented as mean tissue viability compared to negative control (NC – PBS) after 1 h contact with subsequent 42 h post-treatment incubation. Positive control used in the study was 5% SDS. Reference sample (REF) was a mixture of parabens (methylparaben, ethylparaben, propylparaben, butylparaben).

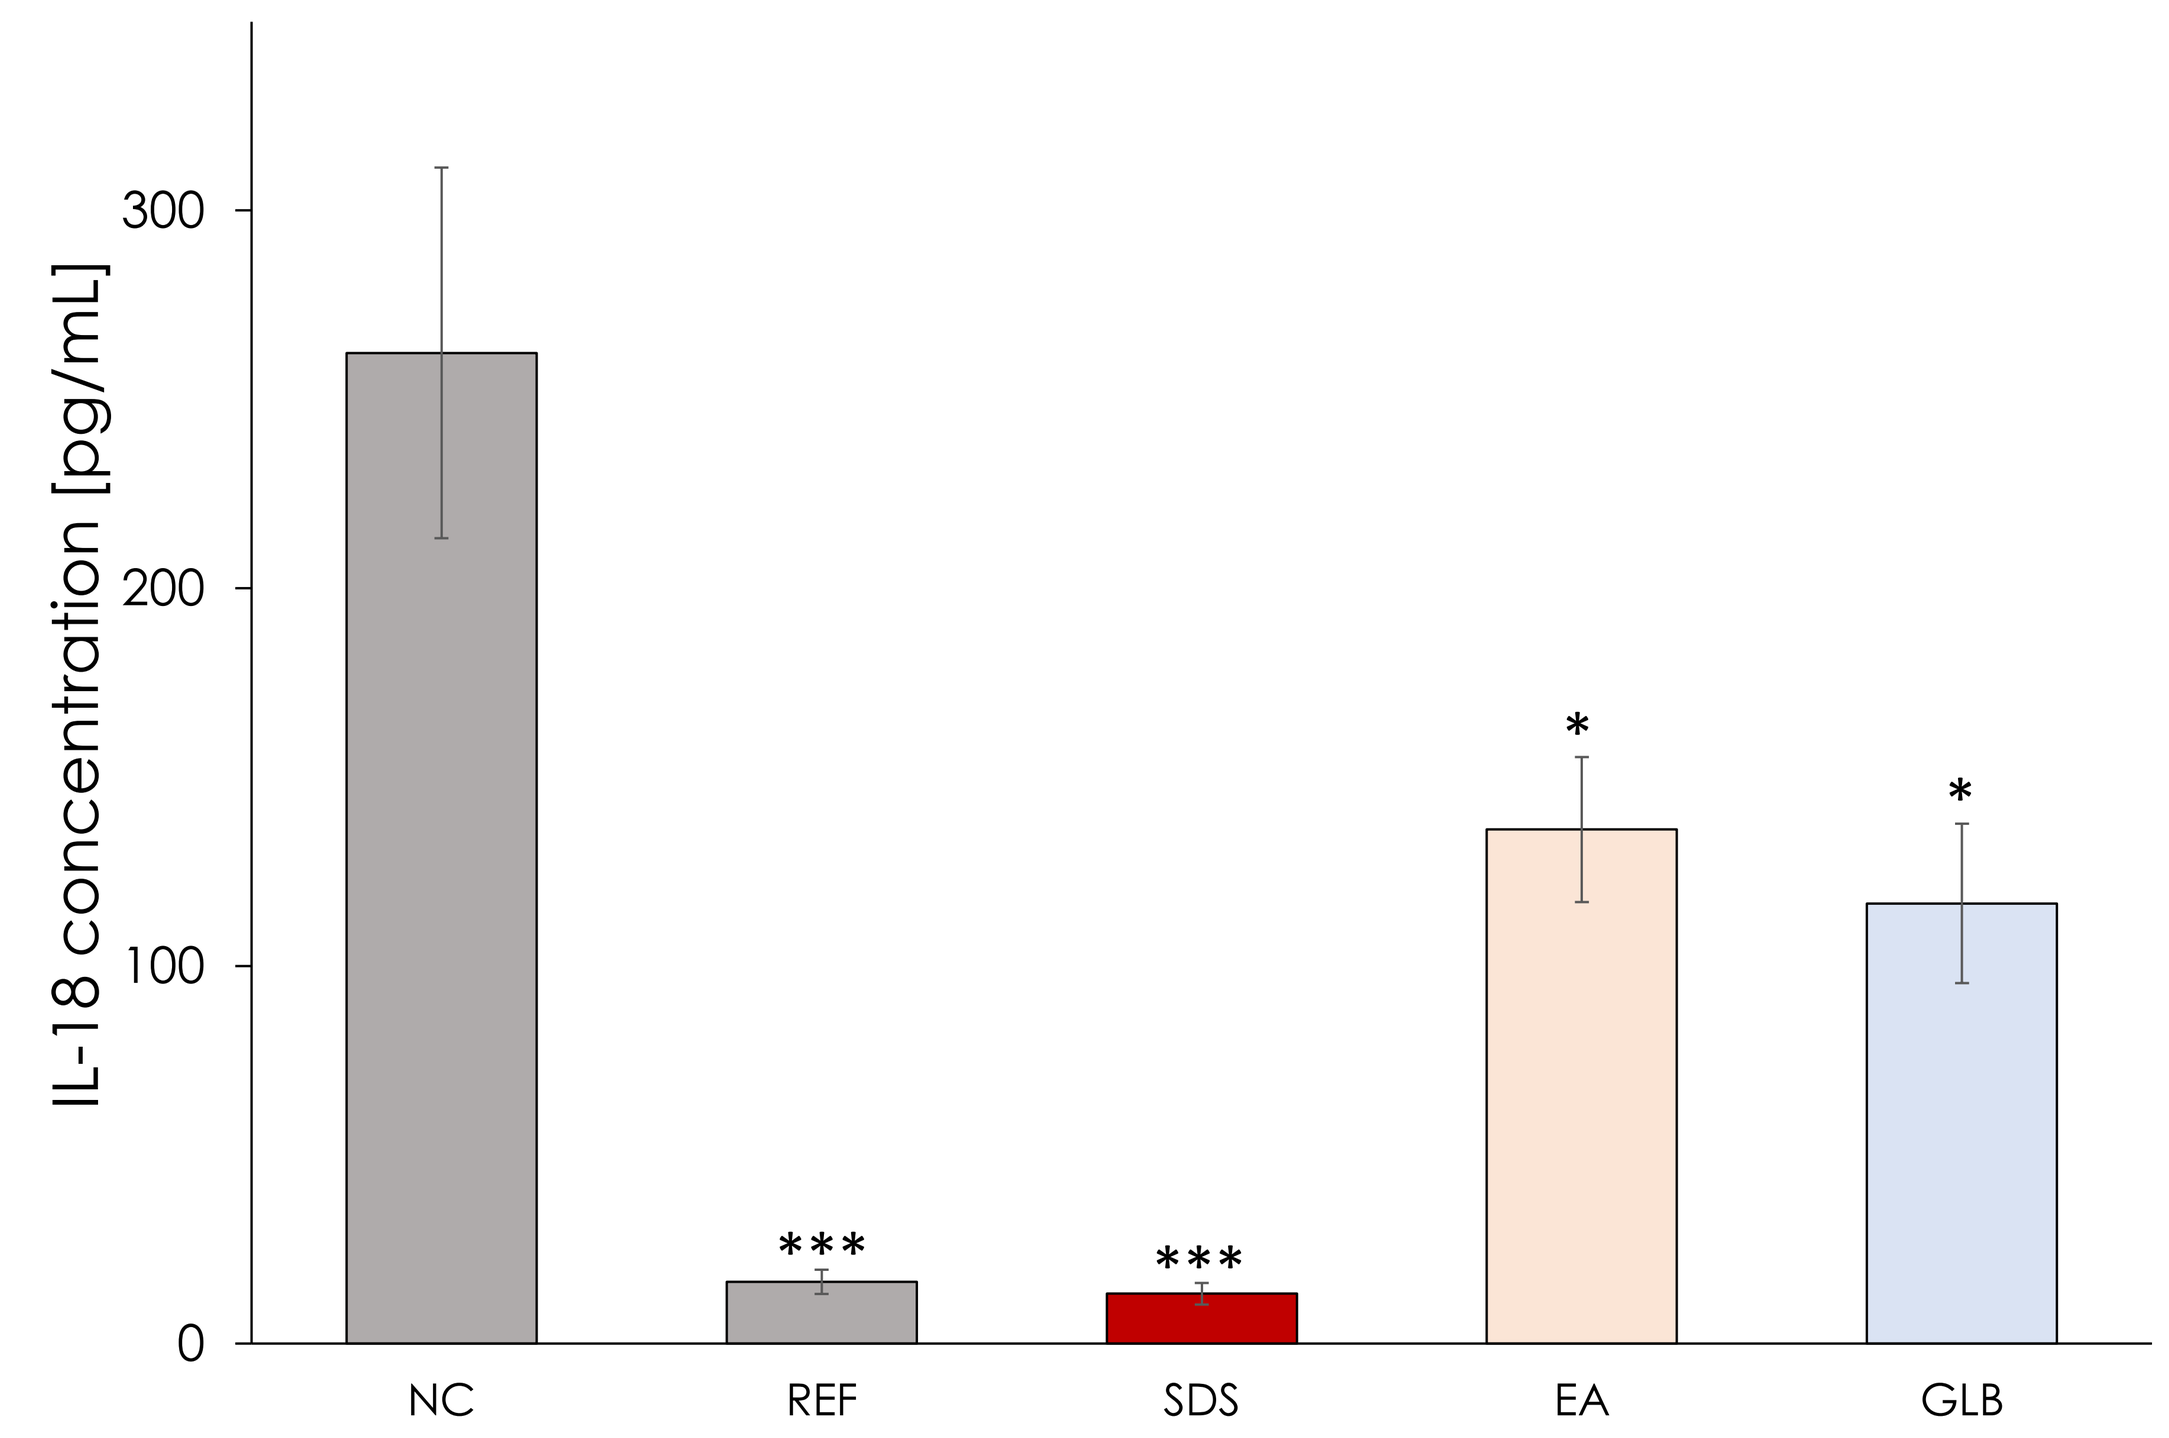
IL-18 concentration [pg/mL]
0
100
200
300

*
*
NC
REF
SDS
EA
GLB

Figure 2. *In vitro* skin sensitization potential of ellagic acid (EA) and glabridin (GLB). Results are presented as mean Interleukin-18 concentration in culture medium after 60-minute direct contact with ellagic acid (EA) and glabridin (GLB), and 24-hour subsequent incubation. Results are presented as mean from 3 individual tissues tested in duplicate (*n*=6) Negative control (NC) used in the study was PBS. Positive control was 5% SDS. Reference sample (REF) was a mixture of parabens (methylparaben, ethylparaben, propylparaben, butylparaben). Statistical significance was calculated using Student's *t*-test (* (*p*<0.05), and *** (*p*<0.01)).

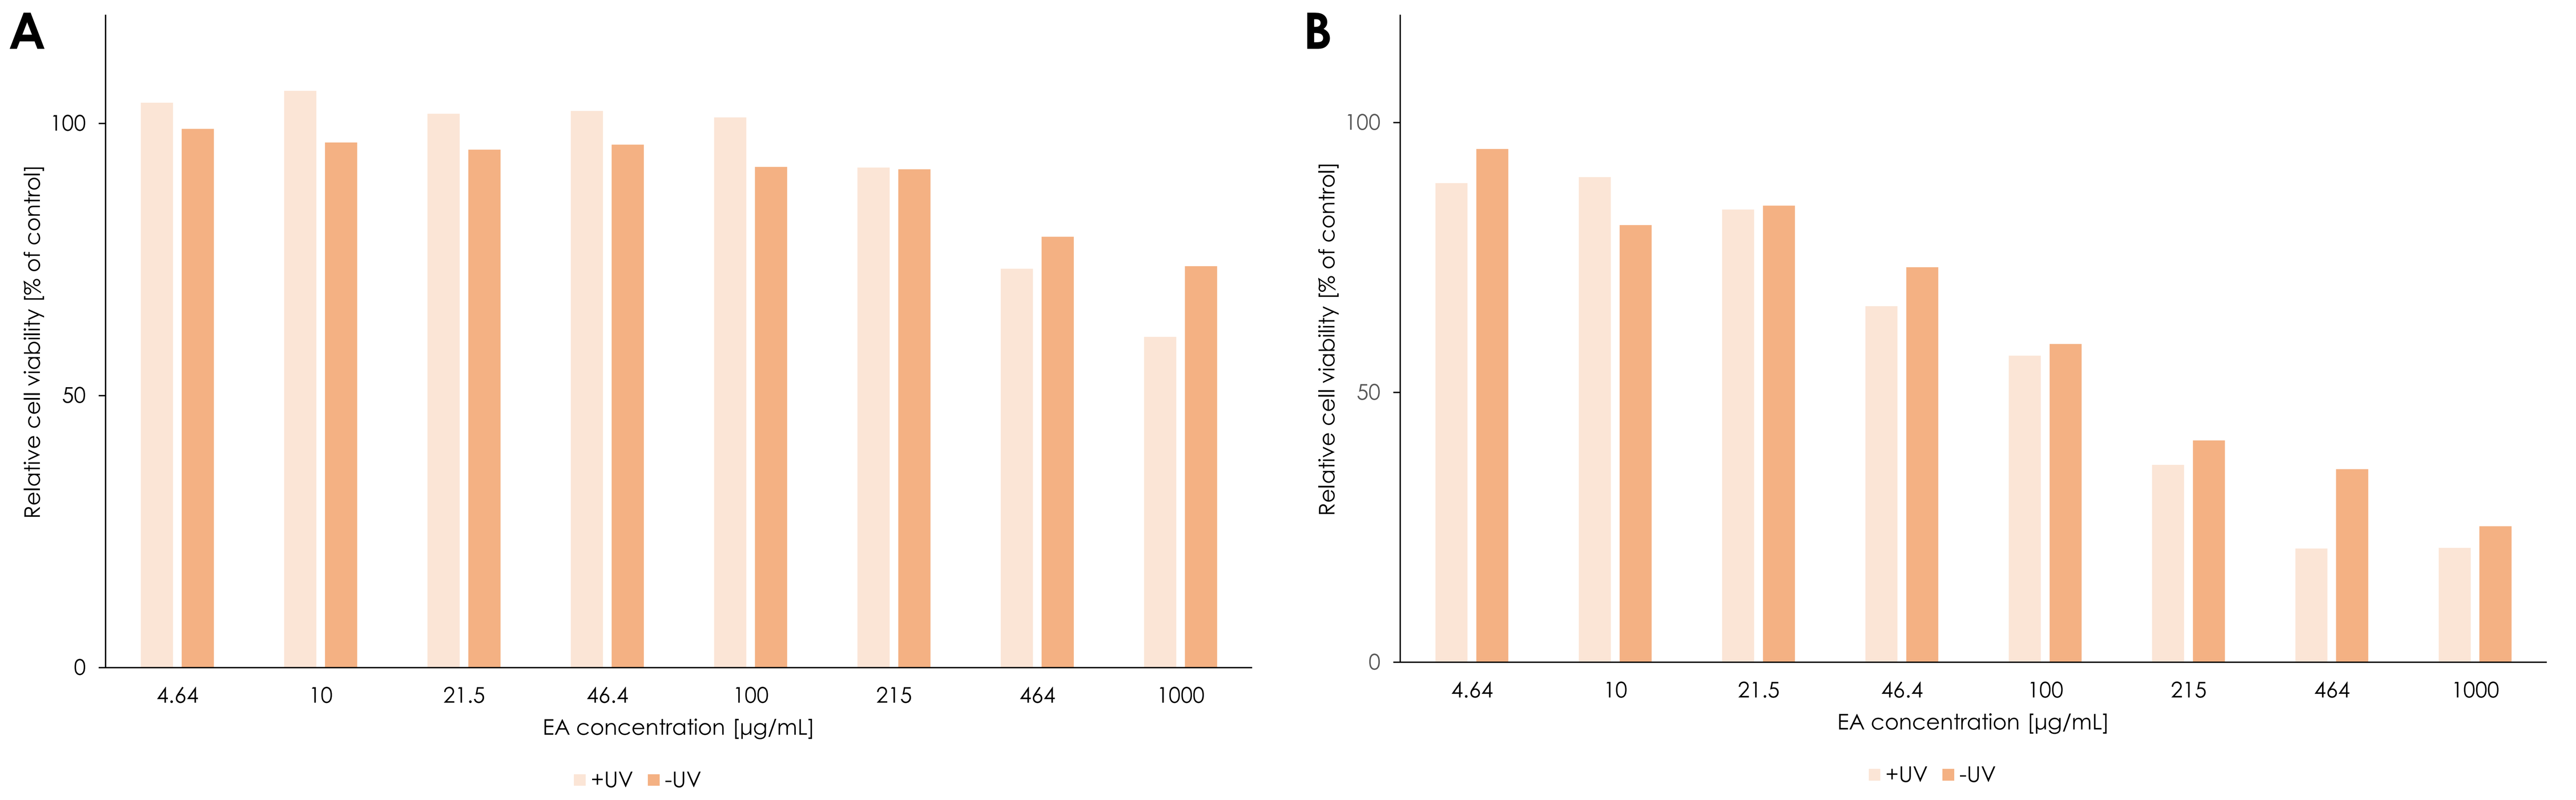
A
B
Relative cell viability [% of control]
0
50
100
4.64
10
21.5
46.4
100
215
464
1000
EA concentration [µg/mL]
+UV
-UV

A

	+UV	-UV
IC50 [µg/mL]	>1000 µg/mL	>1000 µg/mL

B

	+UV	-UV
IC50 [µg/mL]	111.8	165.0

Figure 3. *In vitro* phototoxicity analysis of ellagic acid (EA). Results are presented as mean Balb/c 3T3 cell viability referred to as percentage of viability in negative control (untreated cells). EA was dissolved in two solvents: PBS (**A**) and DMSO (**B**). Cells were incubated with EA in presence (+UV; 5 J/cm^2) or absence of UV (-UV). Phototoxicity is observed when the IC50 value for irradiated cells is significantly lower than for non-irradiated cells or when the viability of irradiated cells is lower at the same concentrations of the tested chemical than it is in case of non-irradiated cells.

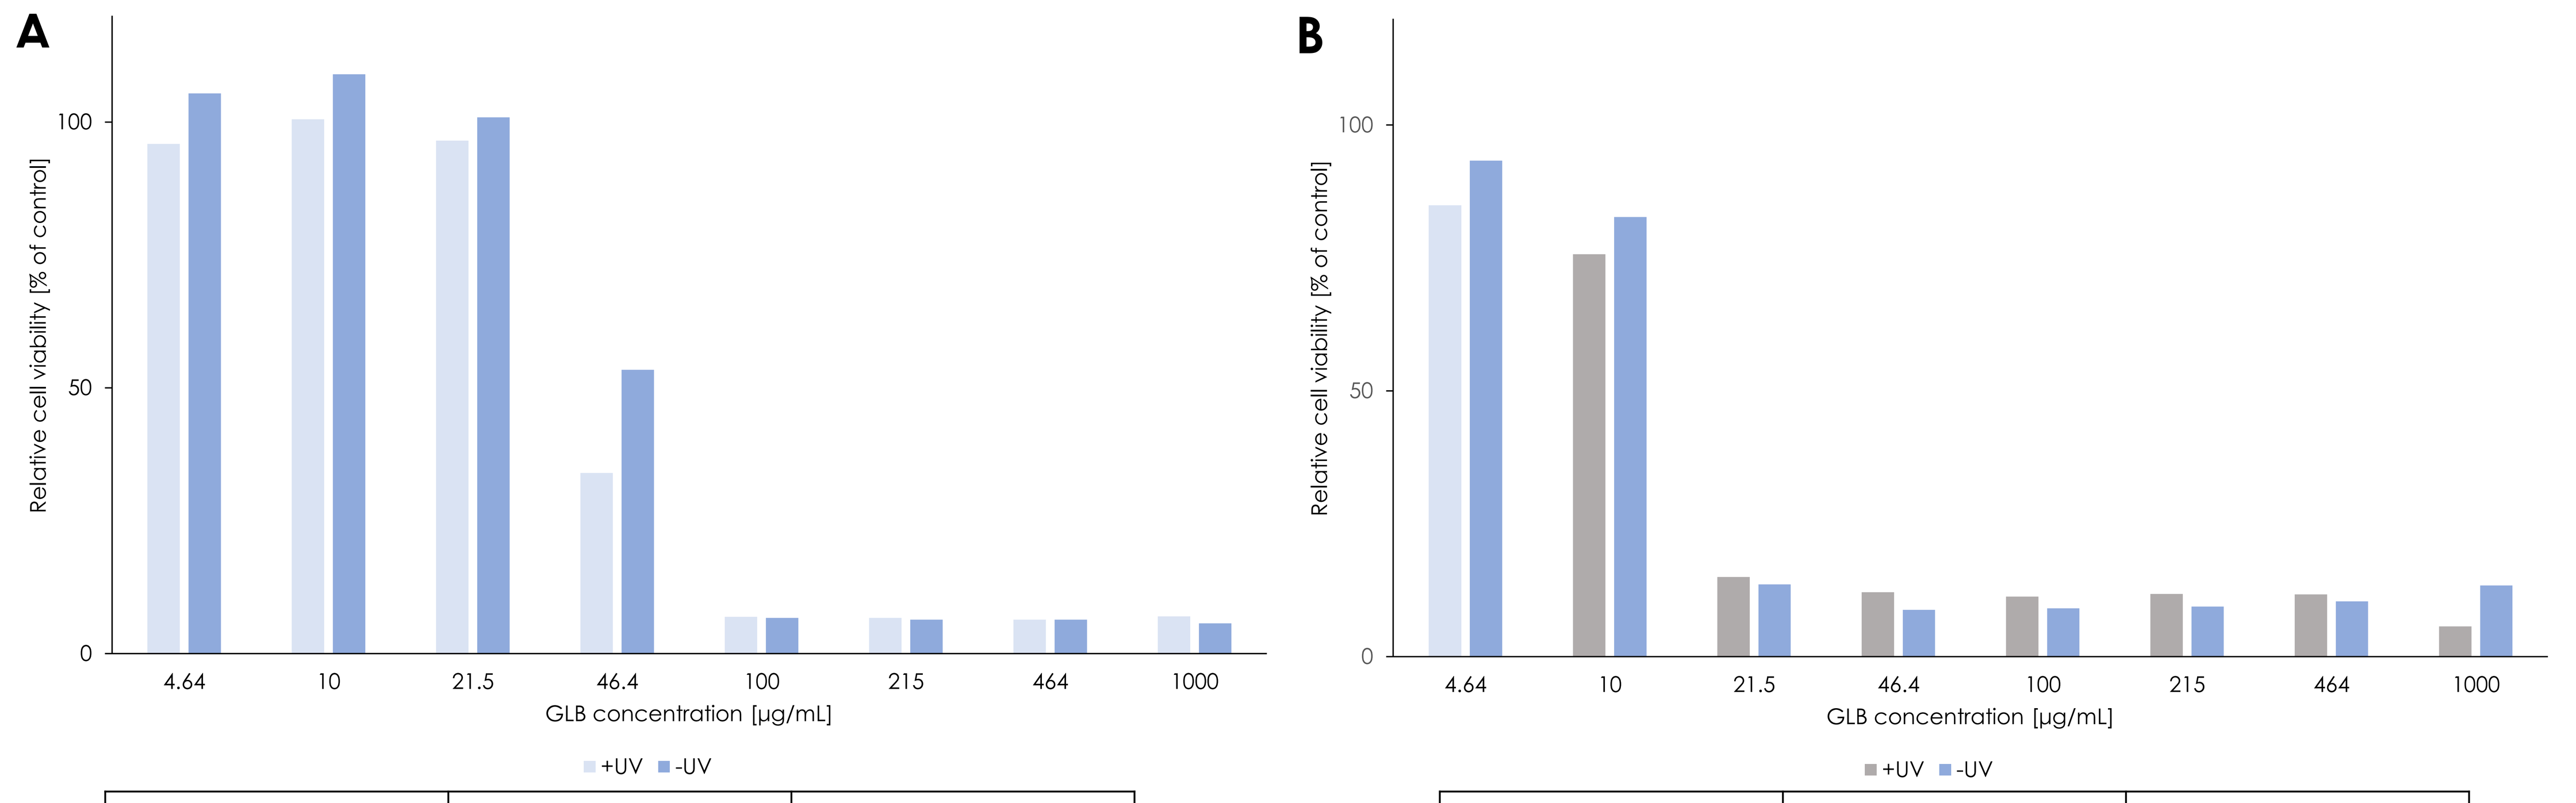
A
B
Relative cell viability [% of control]
0
50
100
4.64
10
21.5
46.4
100
215
464
1000
GLB concentration [µg/mL]
+UV
-UV

A

	+UV	-UV
IC50 [µg/mL]	42.9	47.05

B

	+UV	-UV
IC50 [µg/mL]	14.08	14.77

Figure 4. *In vitro* phototoxicity analysis of glabridin (GLB). Results are presented as mean Balb/c 3T3 cell viability referred to as percentage of viability in negative control (untreated cells). EA was dissolved in two solvents: PBS (**A**) and DMSO (**B**). Cells were incubated with EA in presence (+UV; 5 J/cm^2) or absence of UV(-UV). Phototoxicity is observed when the IC50 value for irradiated cells is significantly lower than for non-irradiated cells or when the viability of irradiated cells is lower at the same concentrations of the tested chemical than it is in case of non-irradiated cells.

Product	Number of volunteers	Adverse events (% of the group)	Notes
Day cream 14711	70 (including 10 pregnant and breastfeeding women)	1(1.43%)	Comedogenic effect after 10 days of using
Night cream 14712	80 (including 10 pregnant and breastfeeding women)	1 (1.25%)	Sebum overproduction in acne-prone area, product well tolerated in other areas
Eye cream 14713	57 (including 10 pregnant and breastfeeding women)	2 (3.51%)	Eye irritation, itching

Table 1. Tolerance of day, night, and eye creams containing glabridin and ellagic acid in volunteers, including pregnant and breastfeeding women, and patients after aesthetic medicine treatments. **Among volunteers, a total of 4 adverse events (AE) occured in the studies. No adverse events were observed in the group of pregnant or breastfeeding women, nor in participants after medical treatments.**

Product	Number of volunteers	Skin reaction after 48/72 hours (% of the group)
Day cream 14711	50 (including 20 volunteers in lactic acid patch test)	0/0 (0%/0%)
Night cream 14712	50 (including 20 volunteers in lactic acid patch test)	0/0 (0%/0%)
Eye cream 14713	30	0/0 (0%/0%)

Table 2. Local skin tolerance of topical formulations containing ellagic acid and glabridin. Patch testing for day cream and night cream included groups whose skin was previously irritated with 10% lactic acid to confirm skin sensitivity. **None of the test subjects developed skin reaction after 48/72 hours of exposure.**

Product	Mean erythema reduction in the whole group (t = 28 days)	Erythema reduction (volunteers with improvement)
Day cream 14711	−13%	**−17%** in 77% of volunteers
Night cream 14712	−11%	**−16%** in 73% volunteers
Eye cream 14713	−10%	**−15%** in 78% volunteers

Table 3. Erythema reduction after 4 weeks of products usage. Measurements of hemoglobin content in the skin (Mexameter MX18) are presented as mean change from the baseline (*mean erythema reduction*) in the whole group and in the group where improvement was observed (*volunteers with improvement*).

CONCLUSIONS

Our observations showed that pure ingredients, as well as their combination may be safely used in topical formulation. *In vitro* studies confirm that products do not induce phototoxicity, nor do they exhibit irritant or sensitizing potential. Topical use of products containing evaluated polyphenols were not only well tolerated by test subjects, but also able to reduce erythema in volunteers' skin.